Treating glioblastoma with a cytokine inhibitor, ibudilast in combination with temozolomide extends survival in a patient xenograft model

K.L. McDonald[1], W, Ha[1], H. Sevim[1], K. Matsuda[2], M. Khasraw[3]

1. Cure Brain Cancer Foundation Biomarkers and Translational Research, Prince of Wales Clinical School, University of NSW, Australia; 2. MediciNova Inc. La Jolla, CA, USA; 3. NHMRC Clinical Trials Centre, University of Sydney, Australia

Introduction

The standard treatment for newly diagnosed glioblastoma patients consists of concurrent radiotherapy and temozolomide followed by adjuvant temozolomide. The methylation of *MGMT* confers significant survival benefit to this treatment regime. However, despite their better prognosis, resistance to treatment is acquired and the tumour recurs.

The Aims of this study were to:

(1) Compare the proteomic profiles of *MGMT* methylated glioblastoma samples when grouped according to survival (±1 year)
(2) Identify novel biomarkers associated with poor survival
(3) Further explore these biomarkers as potential candidates for treatment.

Mass Spectrometry

Frozen tissue from 30 MGMT methylated glioblastomas were identified for proteomic profiling using SELDI-TOF mass spectrometry.

Patients were grouped according to survival:
Group 1: Poor survivors defined by survival of less than 12 months
Group 2: Good survivors defined by survival of more than 12 months

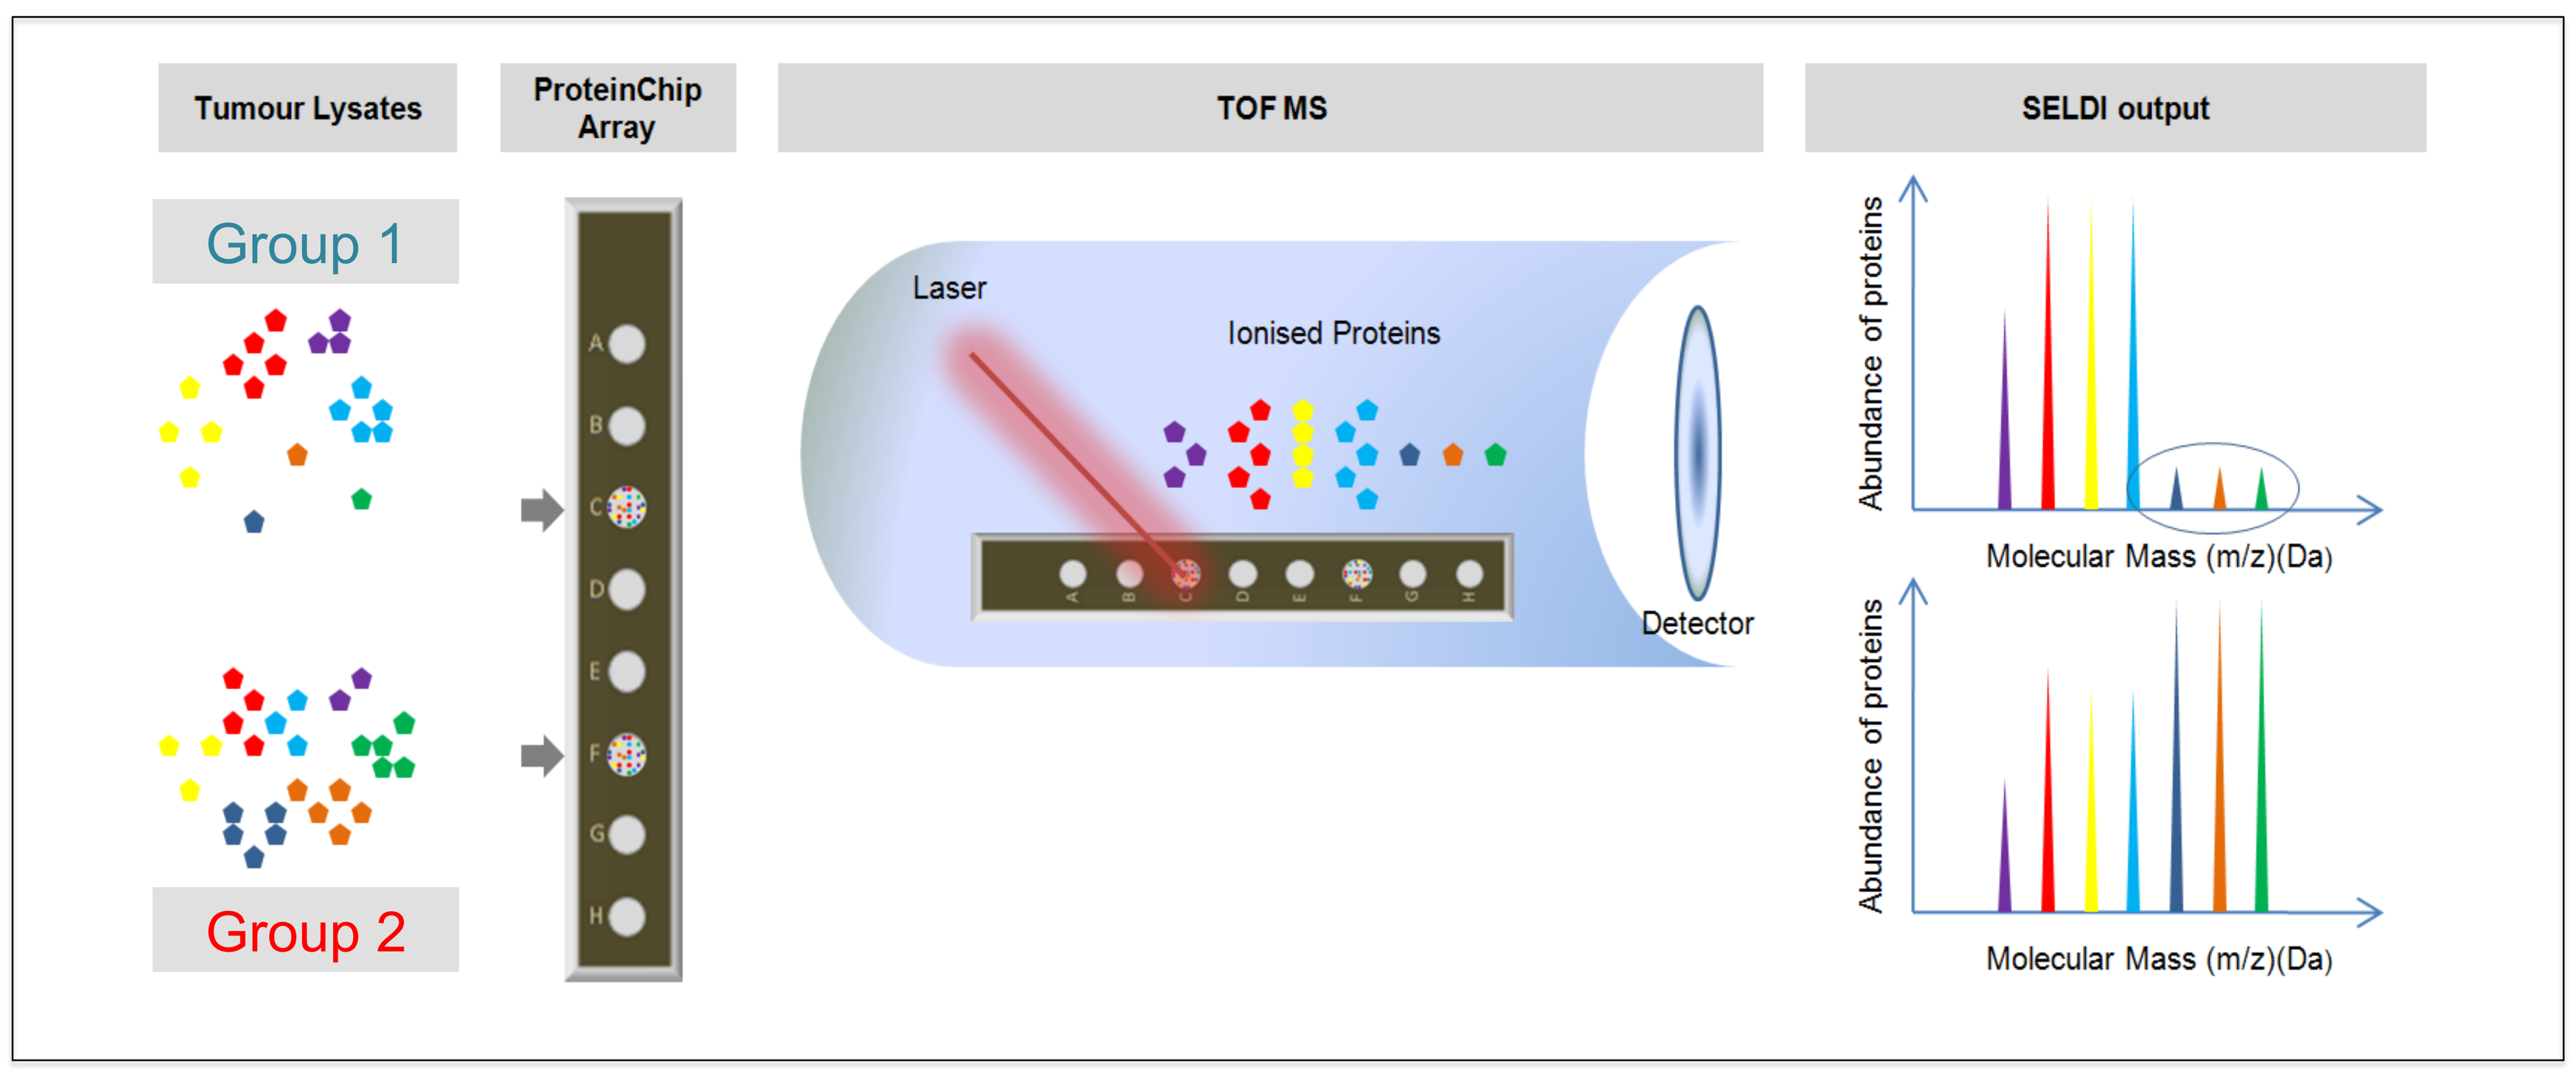
Tumour Lysates
ProteinChip Array
TOF MS
SELDI output
Group 1
Group 2
Laser
Ionised Proteins
Detector
Abundance of proteins
Molecular Mass (m/z)(Da)

Macrophage Inhibitory Factor (MIF)

A 12.5 kDa protein specifically expressed in the "poor responders" was identified to be Macrophage Inhibitor Factor (MIF). MIF is an inflammatory-related cytokine secreted by cancer stem cells.

We analyzed expression levels of MIF and its receptor, CD-74 in 168 glioblastoma tissues. Co-expression of MIF and CD-74 were found in 57% of glioblastoma tissues and co-expression was significantly associated with poor overall survival.

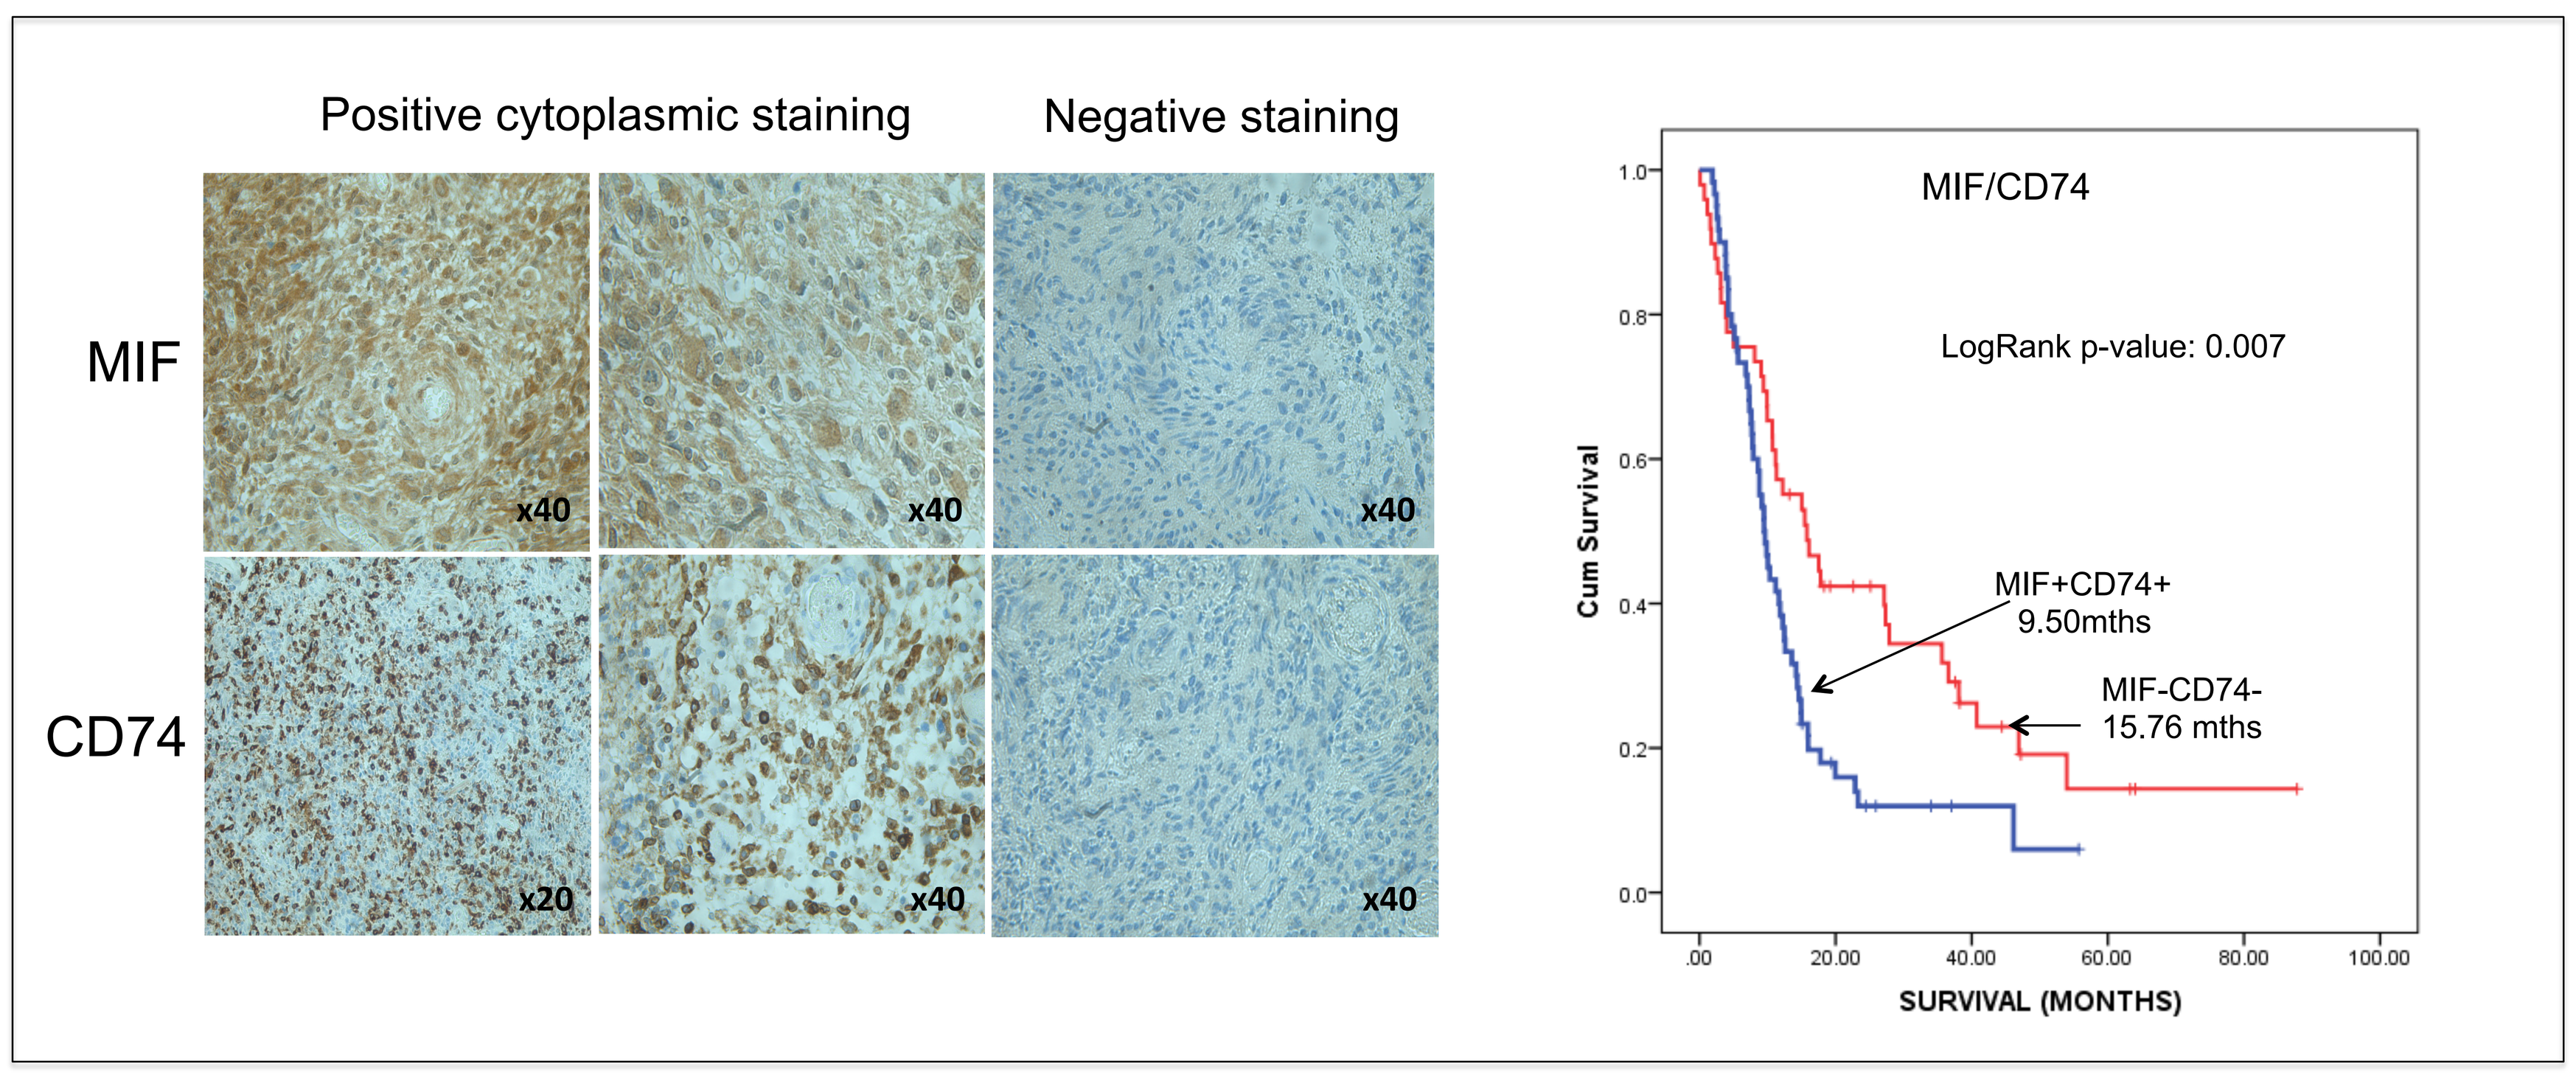
Positive cytoplasmic staining
Negative staining
MIF
CD74
x40
x20
MIF/CD74
LogRank p-value: 0.007
MIF+CD74+ 9.50mths
MIF-CD74- 15.76 mths
Cum Survival
SURVIVAL (MONTHS)

siRNA knockdown of MIF reduces tumour cell proliferation and sensitizes cell to TMZ

We blocked MIF expression using a transient knock-down approach. Cell proliferation was impeded. Significant sensitivity to temozolomide was observed when MIF was blocked.

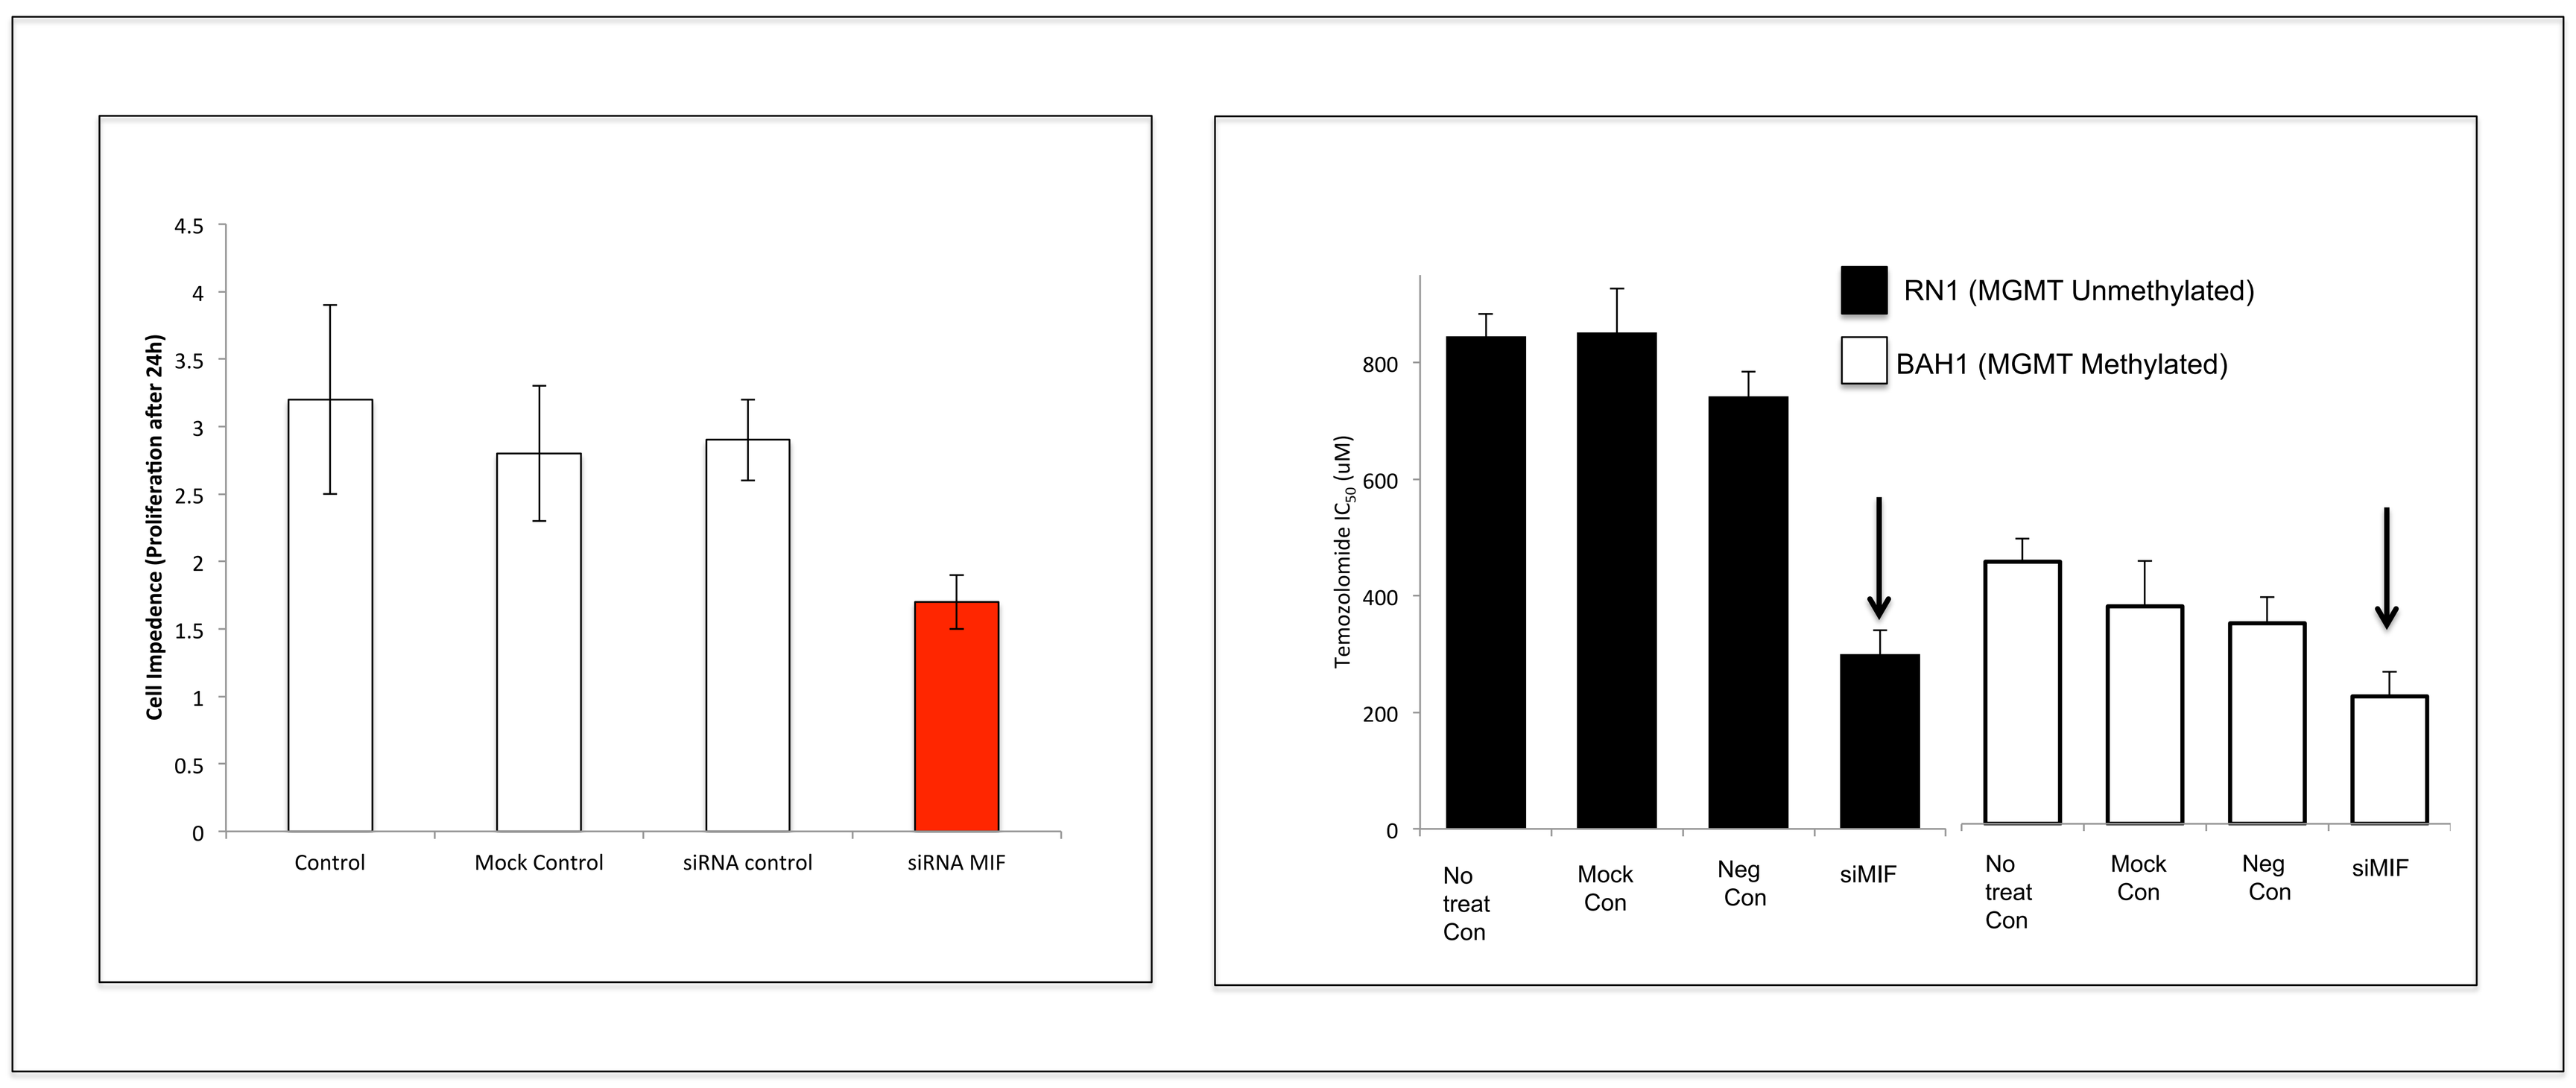
Cell Impedence (Proliferation after 24h)
Control
Mock Control
siRNA control
siRNA MIF
Temozolomide IC50 (uM)
RN1 (MGMT Unmethylated)
BAH1 (MGMT Methylated)
No treat Con
Mock Con
Neg Con
siMIF

Ibudilast (MN-166)

Ibudilast is an orally available, small molecule with anti-inflammatory and neuroprotective effects. It has been available in the Japanese and Korean markets for the treatment of asthma (20 mg/day) and cerebrovascular disorders (30 mg/day) for over 20 years.

Ibudilast is a specific inhibitor of MIF.

We treated a panel of patient-derived cell lines with ibudilast alone and in combination with temozolomide. In each and every cell line tested, we observed significant synergy with ibudilast and temozolomide.

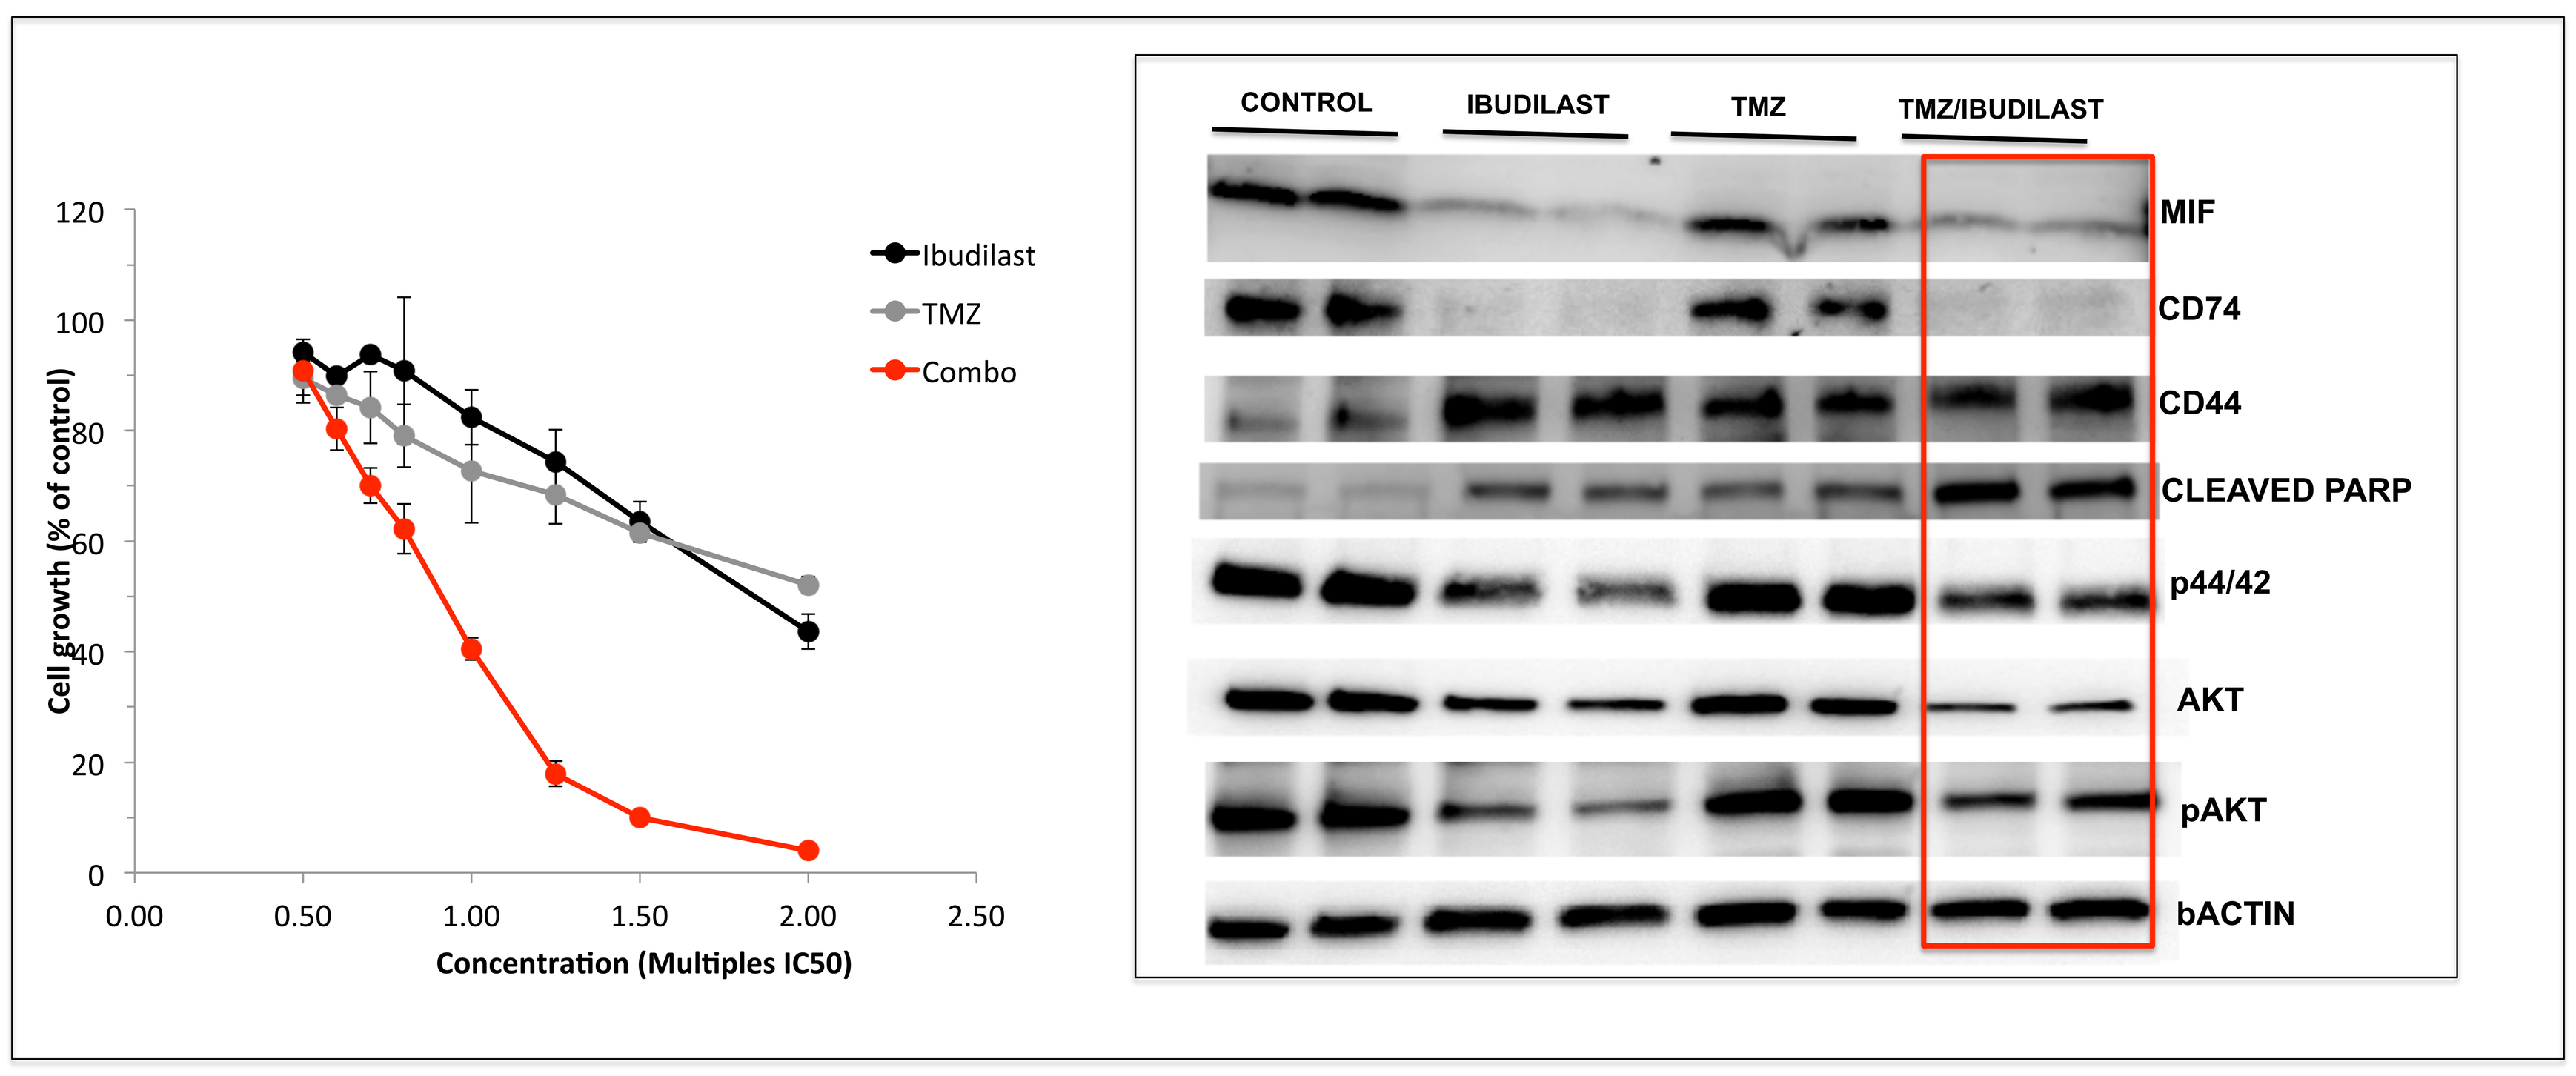
Ibudilast
TMZ
Combo
Cell growth (% of control)
Concentration (Multiples IC50)
CONTROL
IBUDILAST
TMZ
TMZ/IBUDILAST
MIF
CD74
CD44
CLEAVED PARP
p44/42
AKT
pAKT
bACTIN

Combination of ibudilast and temozolomide results in G2 arrest and increased apoptosis

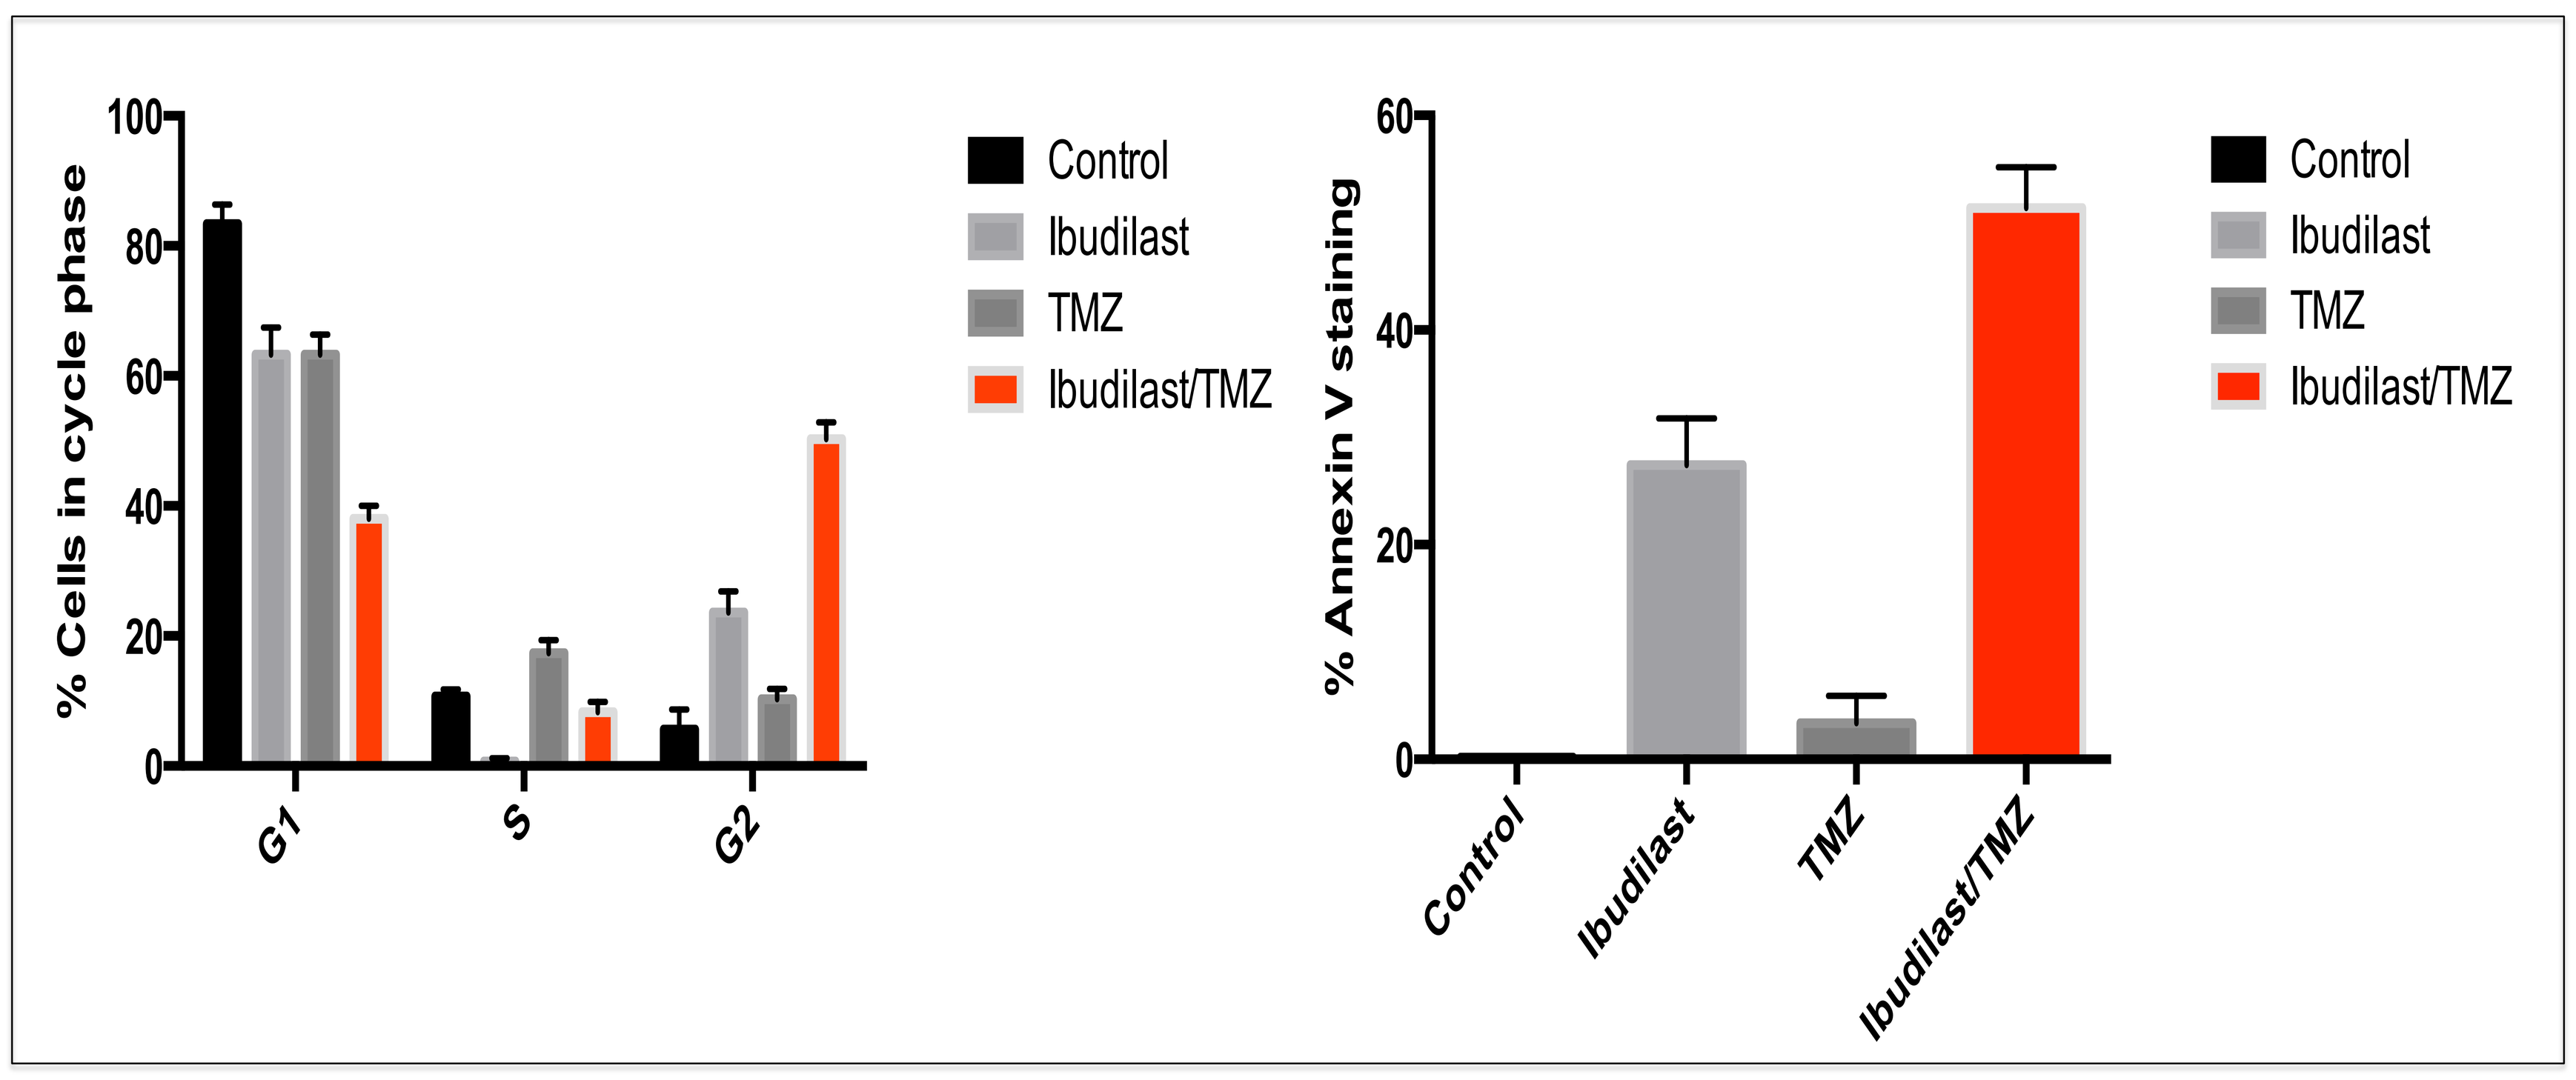
% Cells in cycle phase
G1
S
G2
% Annexin V staining
Control
Ibudilast
TMZ
Ibudilast/TMZ

In vivo, the combination of ibudilast and temozolomide leads to significantly enhanced survival

We tested the combination of ibudilast and temozolomide in our PDX model: RN1 cells (MGMT unmethylated) intracranially implanted into the brains of balb/c nude mice. We administered ibudilast (5mg/kg or 20mg/kg) using oral gavage and temozolomide (10mg/kg) by I.P.

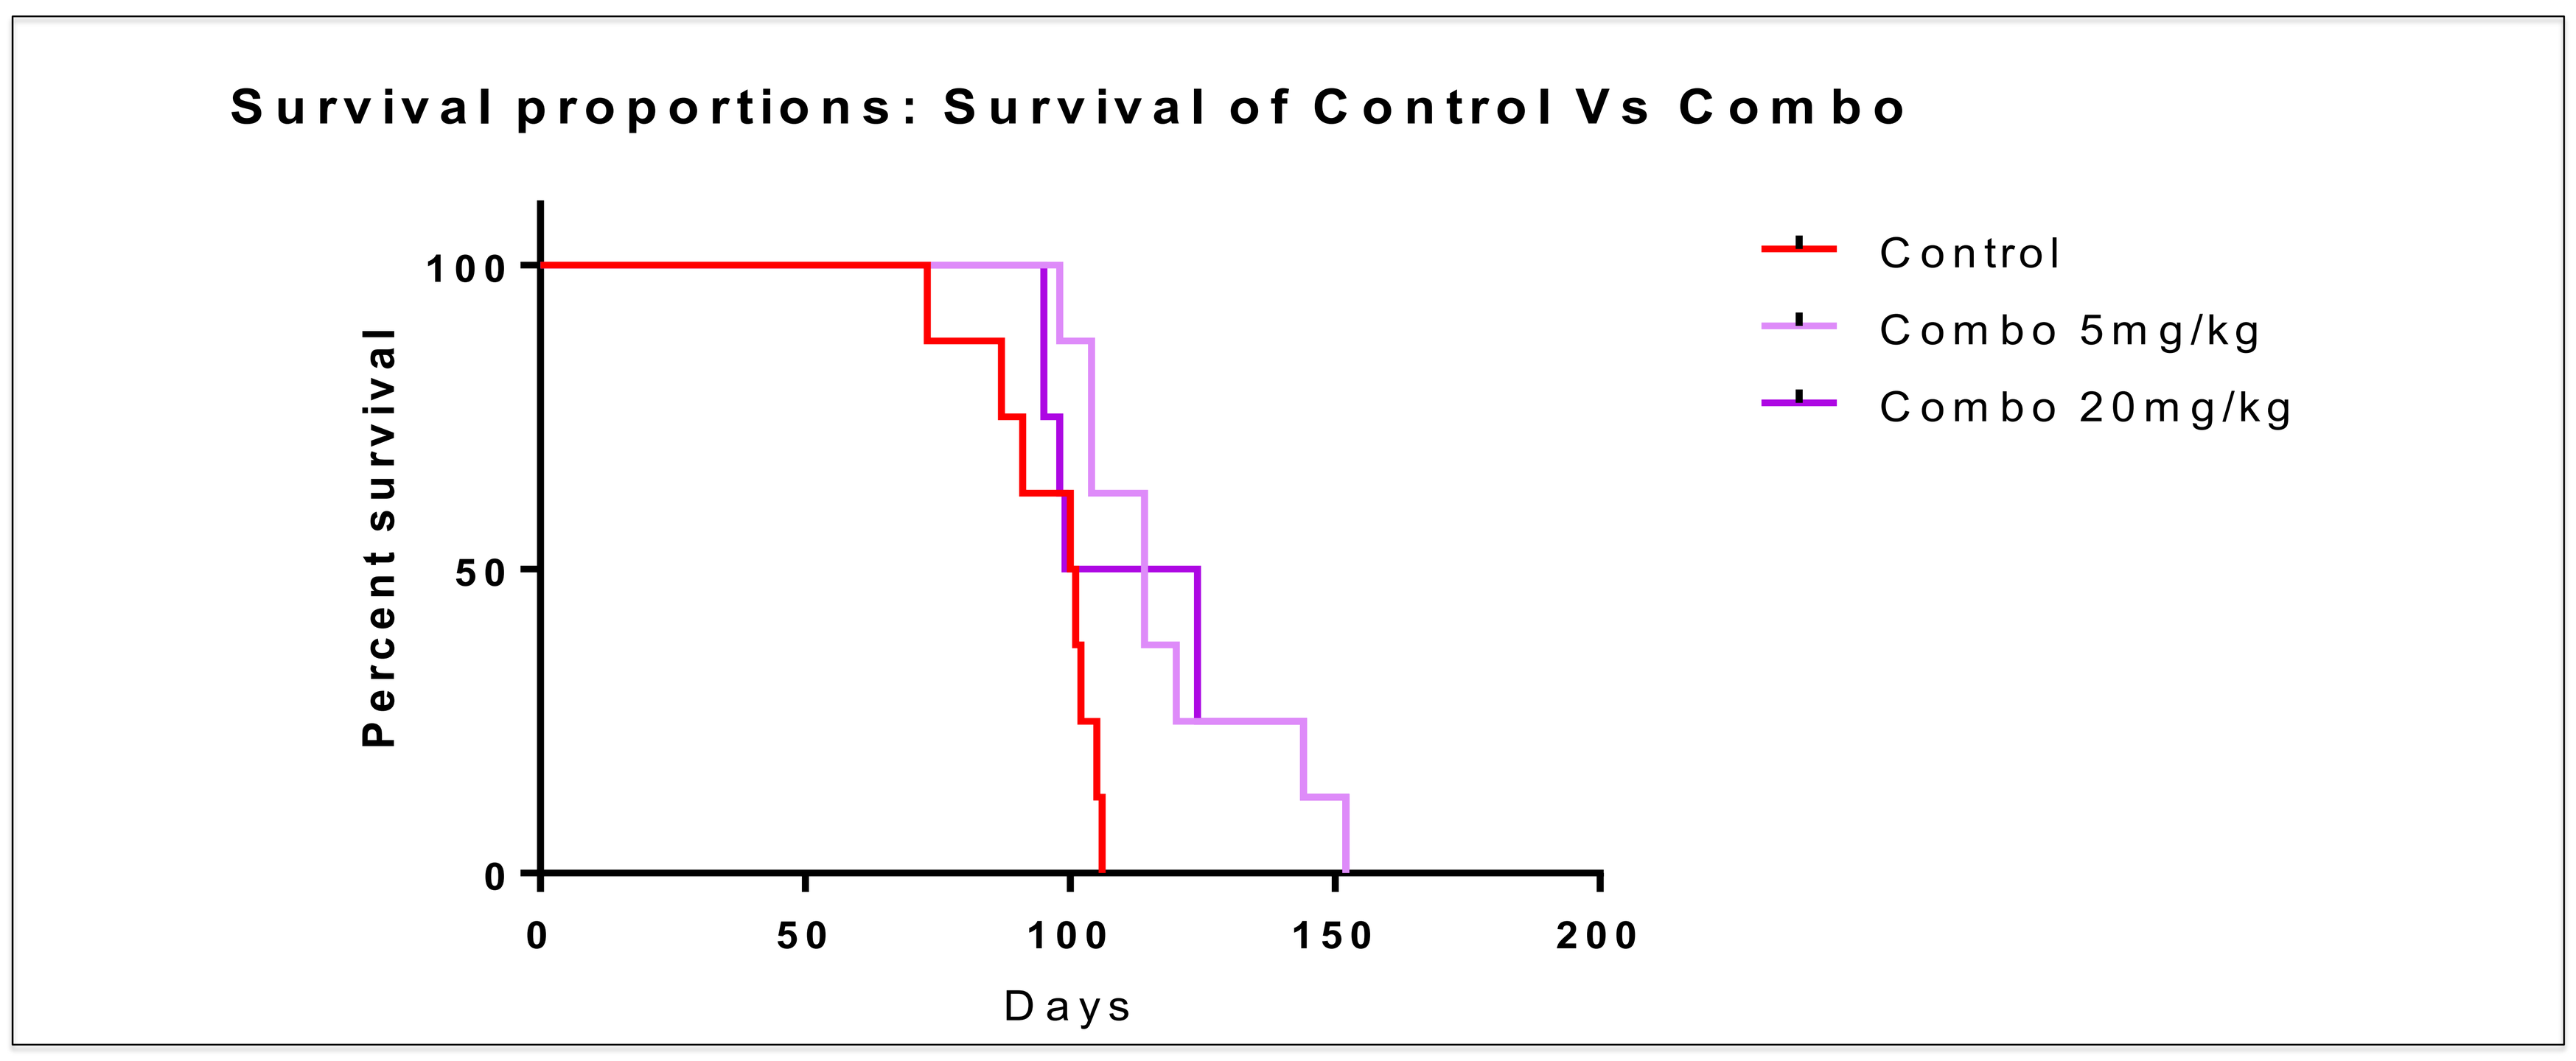
Survival proportions: Survival of Control Vs Combo
Percent survival
Days
Control
Combo 5mg/kg
Combo 20mg/kg

Significantly enhanced survival was observed when mice where treated with combination treatment compared to control mice. Median survival was 114 days (treated with combination) compared to control (100.5 days). Expression levels of MIF and its receptor, CD74 were suppressed.

Conclusions:

The combination of ibudilast and temozolomide leads to improved survival in a patient-derived xenograft model. The combination was well-tolerated.

A phase 1 trial is being planned to assess the efficacy of ibudilast (60mg/kg) combined with temozolomide (5 day schedule) for patients with recurrent glioblastoma.