Ibudilast, a Novel Neuroimmune Modulator, Decreases Alcohol Craving and Increases Positive Mood in an AUD Population

Daniel J.O. Roche, Spencer Bujarski, and Lara A. Ray (ACNP Associate Member)

[1]University of California Los Angeles, Department of Psychology

BACKGROUND

- The development of novel pharmacotherapies for alcohol use disorder (AUD) represents a critical public health objective
- Neuroinflammation may be related to alcohol's acute behavioral effects, alcohol consumption, and alcohol-induced neurotoxicity.
 - Neuroinflammatory processes may represent a novel therapeutic target for AUD
- Ibudilast (IBUD) inhibits phosphodiesterases -4 and -10 and macrophage migration inhibitory factor, thereby attenuating glial activation and pro-inflammatory signaling.
- Recently, IBUD has been shown to reduce alcohol consumption in rats, but its efficacy as a potential AUD pharmacotherapy has not been tested in humans.
- Therefore, the objective of this Phase I/Phase II laboratory study was to determine the safety, tolerability, and initial efficacy of IBUD (50 mg BID) as a potential AUD medication.

METHODS

SUBJECTS & PROCEDURE

- Twenty four non-treatment-seeking individuals with AUD completed 2 separate 6-day medication regimens (50 mg IBUD BID or placebo, in a randomized, counterbalanced, and crossover design) separated by a 5-10 day washout period.
 - IBUD titration was as follows: 20 mg BID on days 1-2 and 50 mg BID on days 3-6.
- Participants completed the following experimental paradigms during each regimen:
 - On day 5, a guided imagery stress exposure.
 - On day 6, an alcohol cue reactivity task followed by an alcohol infusion paradigm.
 - Alcohol infusion: a 6% ethanol solution was administered via IV infusion. At each target BrAC, i.e. 0.020 g/dl, 0.040g/dl, 0.060g/dl, and 0.080g/dl, the infusion rate was halved, and participants then completed a battery of subjective measures.
 - After the infusion, participants remained in the UCLA CTRC overnight and were discharged on day 7.

MEASURES

- Alcohol craving (AUQ) and adverse effects (SAFTEE) were assessed on a daily basis.
- Mood (POMS) and alcohol craving (AUQ) measures were assessed during the cue reactivity and stress procedures.
- Salivary cortisol was measured for 30 minutes before and 60 minutes after the stressor.
- Subjective response (BAES, POMS, AUQ) was measured during the alcohol infusion.

Table 1: Sample Characteristics

	Mean (SD) or %
Age	31.55 (9.25)
Sex - % Male	72.7%
Ethnicity	
% Caucasian	22.70%
% African American	36.40%
Drinking Days per Month	20.91 (6.10)
Drinks per Drinking Day	6.64 (4.21)
AUD Symptom Count	4.86 (2.61)
% Mild AUD	28.57%
% Moderate AUD	38.10%
% Severe AUD	28.57%

RESULTS

Safety

- IBUD was well tolerated and safe during the study. Of 24 potential adverse drug effects, only headache was reported in significantly greater frequency during the IBUD vs. placebo regimen (4 instances vs. 0 instances).

Daily Craving

- IBUD, but not placebo, significantly decreased basal, daily AUQ craving over the course of the study ($p < 0.05$; **Figure 1**).

Alcohol Cue and Stress Reactivity

- IBUD did not affect cue- and stress- induced AUQ craving.
- However, IBUD, vs. placebo, increased positive mood during both the cue reactivity ($p = 0.07$; **Figure 2**) and stress procedures ($p < 0.05$; **Figure 3**).
- IBUD did not affect cortisol reactivity to the stressor, but IBUD produced a modest increase in overall cortisol level vs. placebo ($p = 0.07$; **Figure 4**).

Alcohol Infusion

- IBUD did not affect subjective response to alcohol during the infusion.

Figure 1: Daily Craving

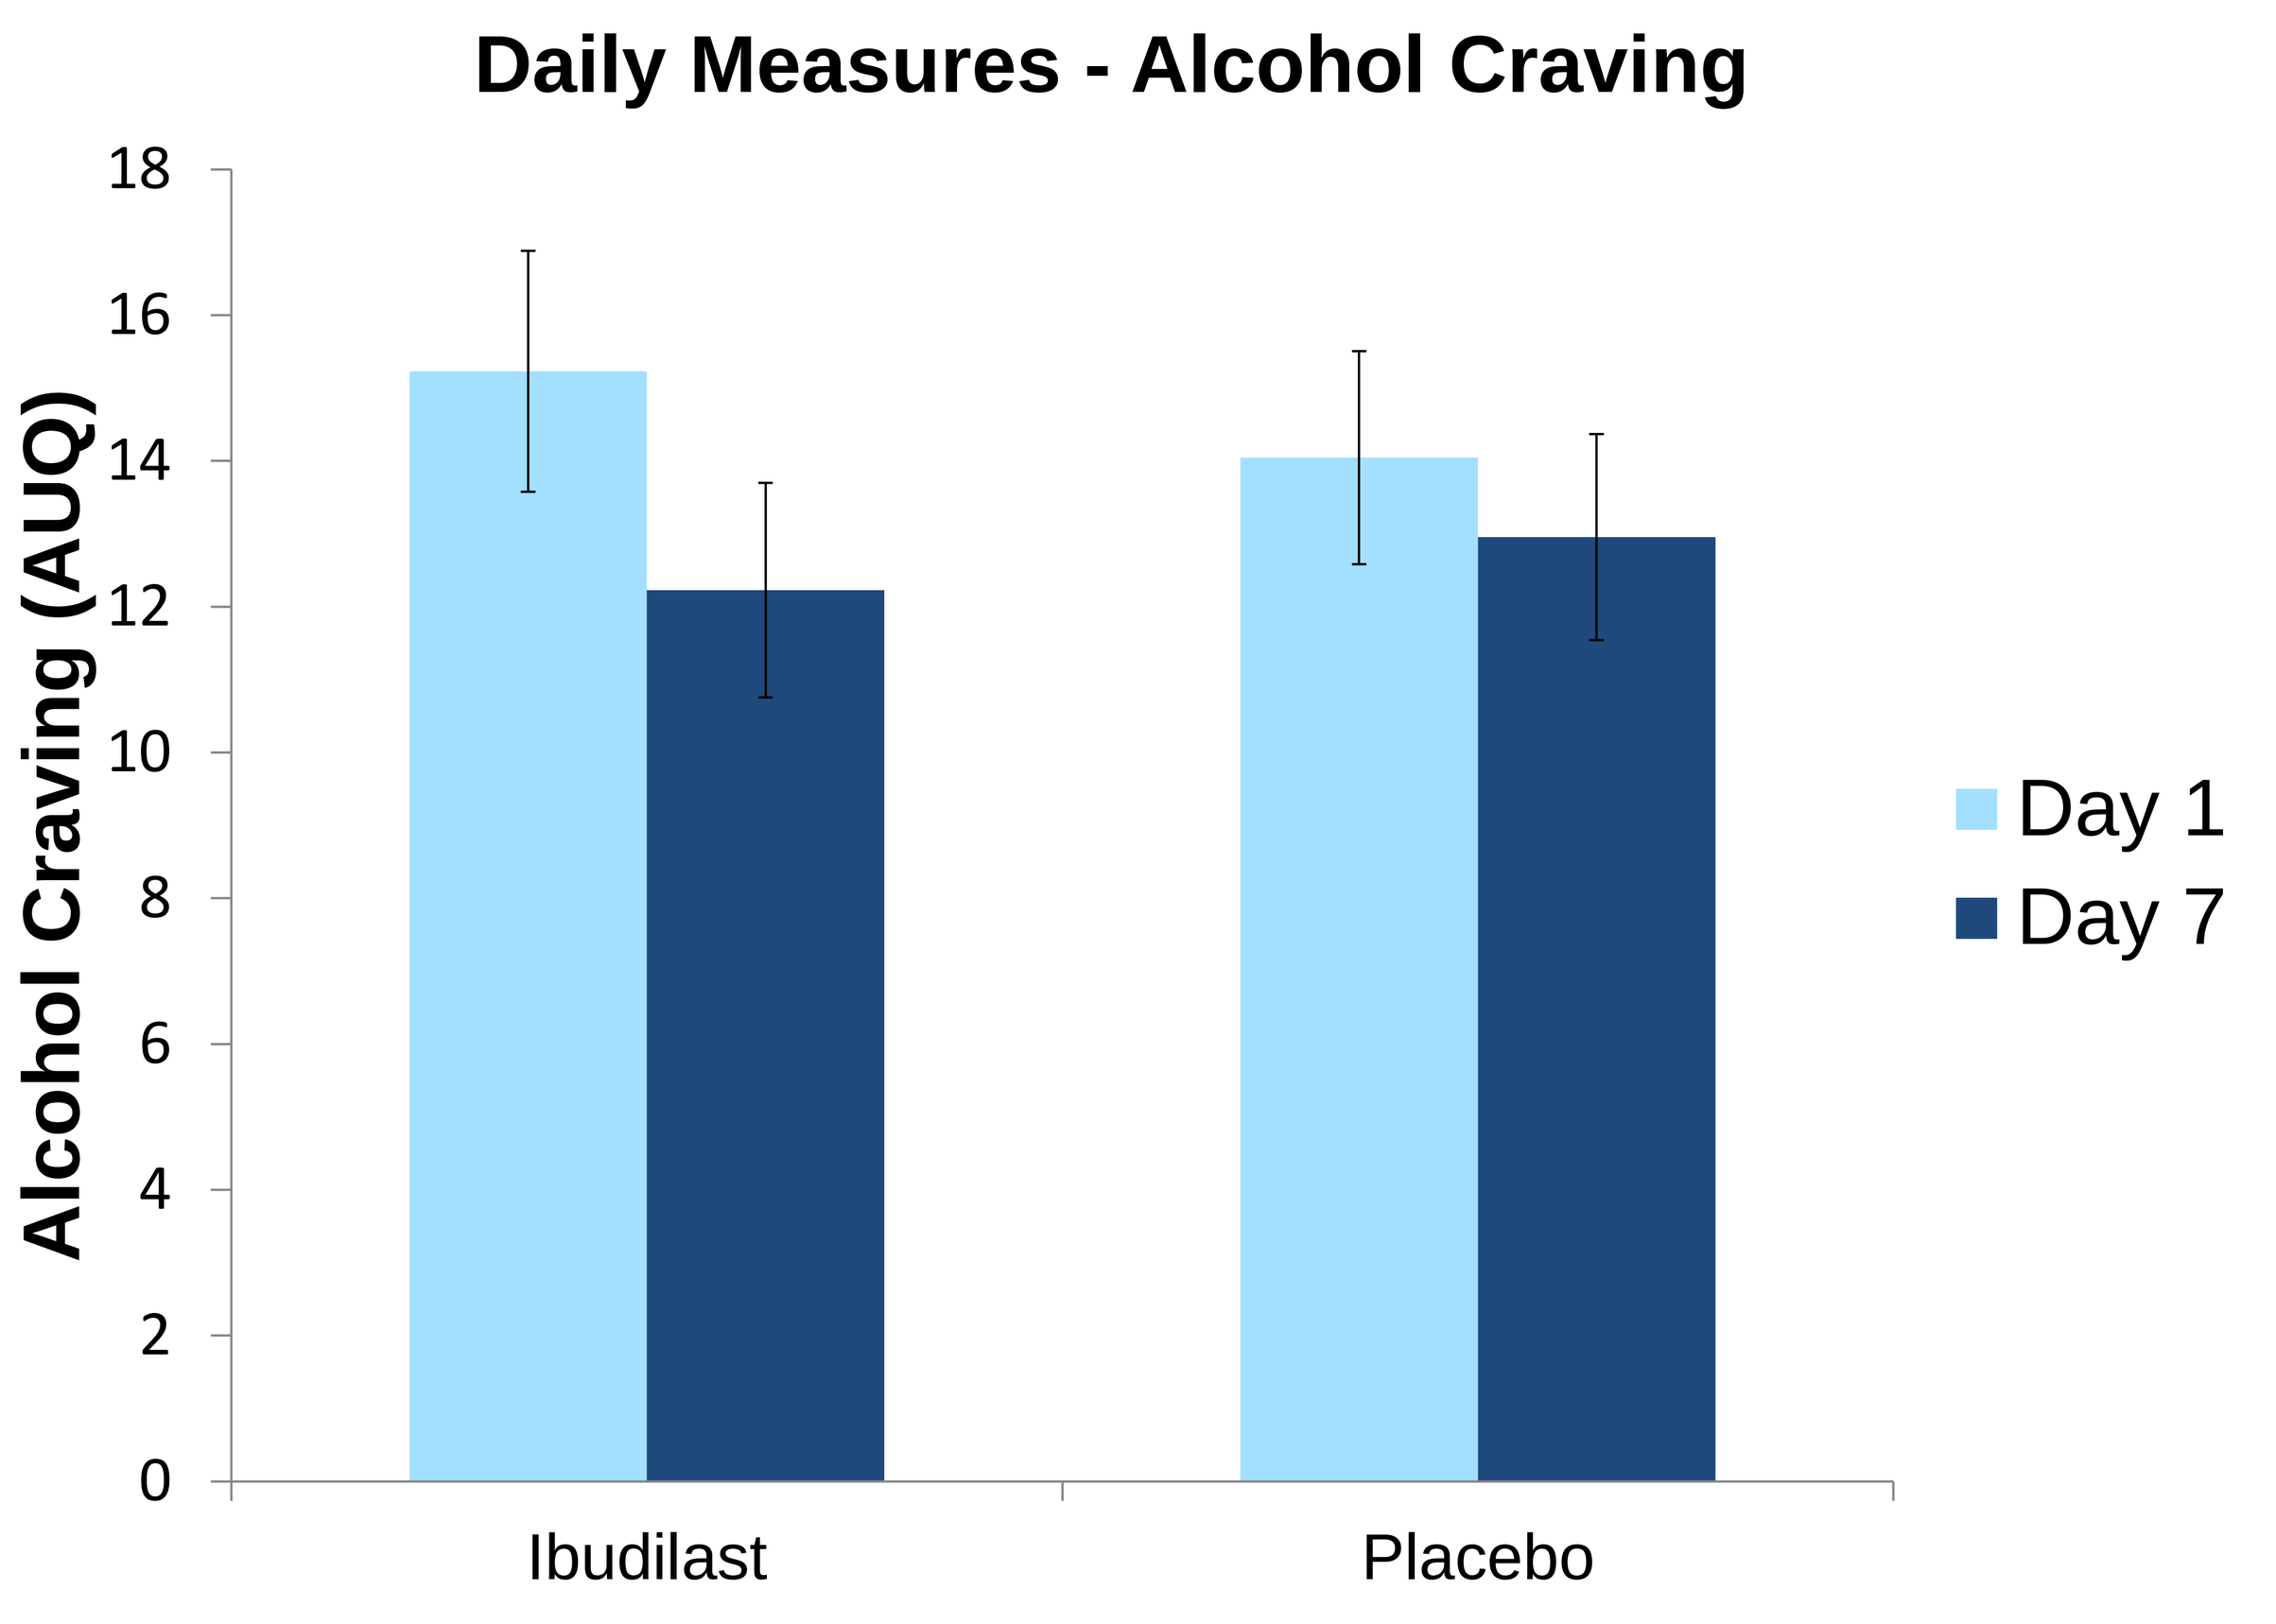
Daily Measures - Alcohol Craving
Alcohol Craving (AUQ)
0
2
4
6
8
10
12
14
16
18
Ibudilast
Placebo
Day 1
Day 7

Medication × Day, $F(1,21) = 4.55$, $p < .05$

Figure 2: Cue Reactivity

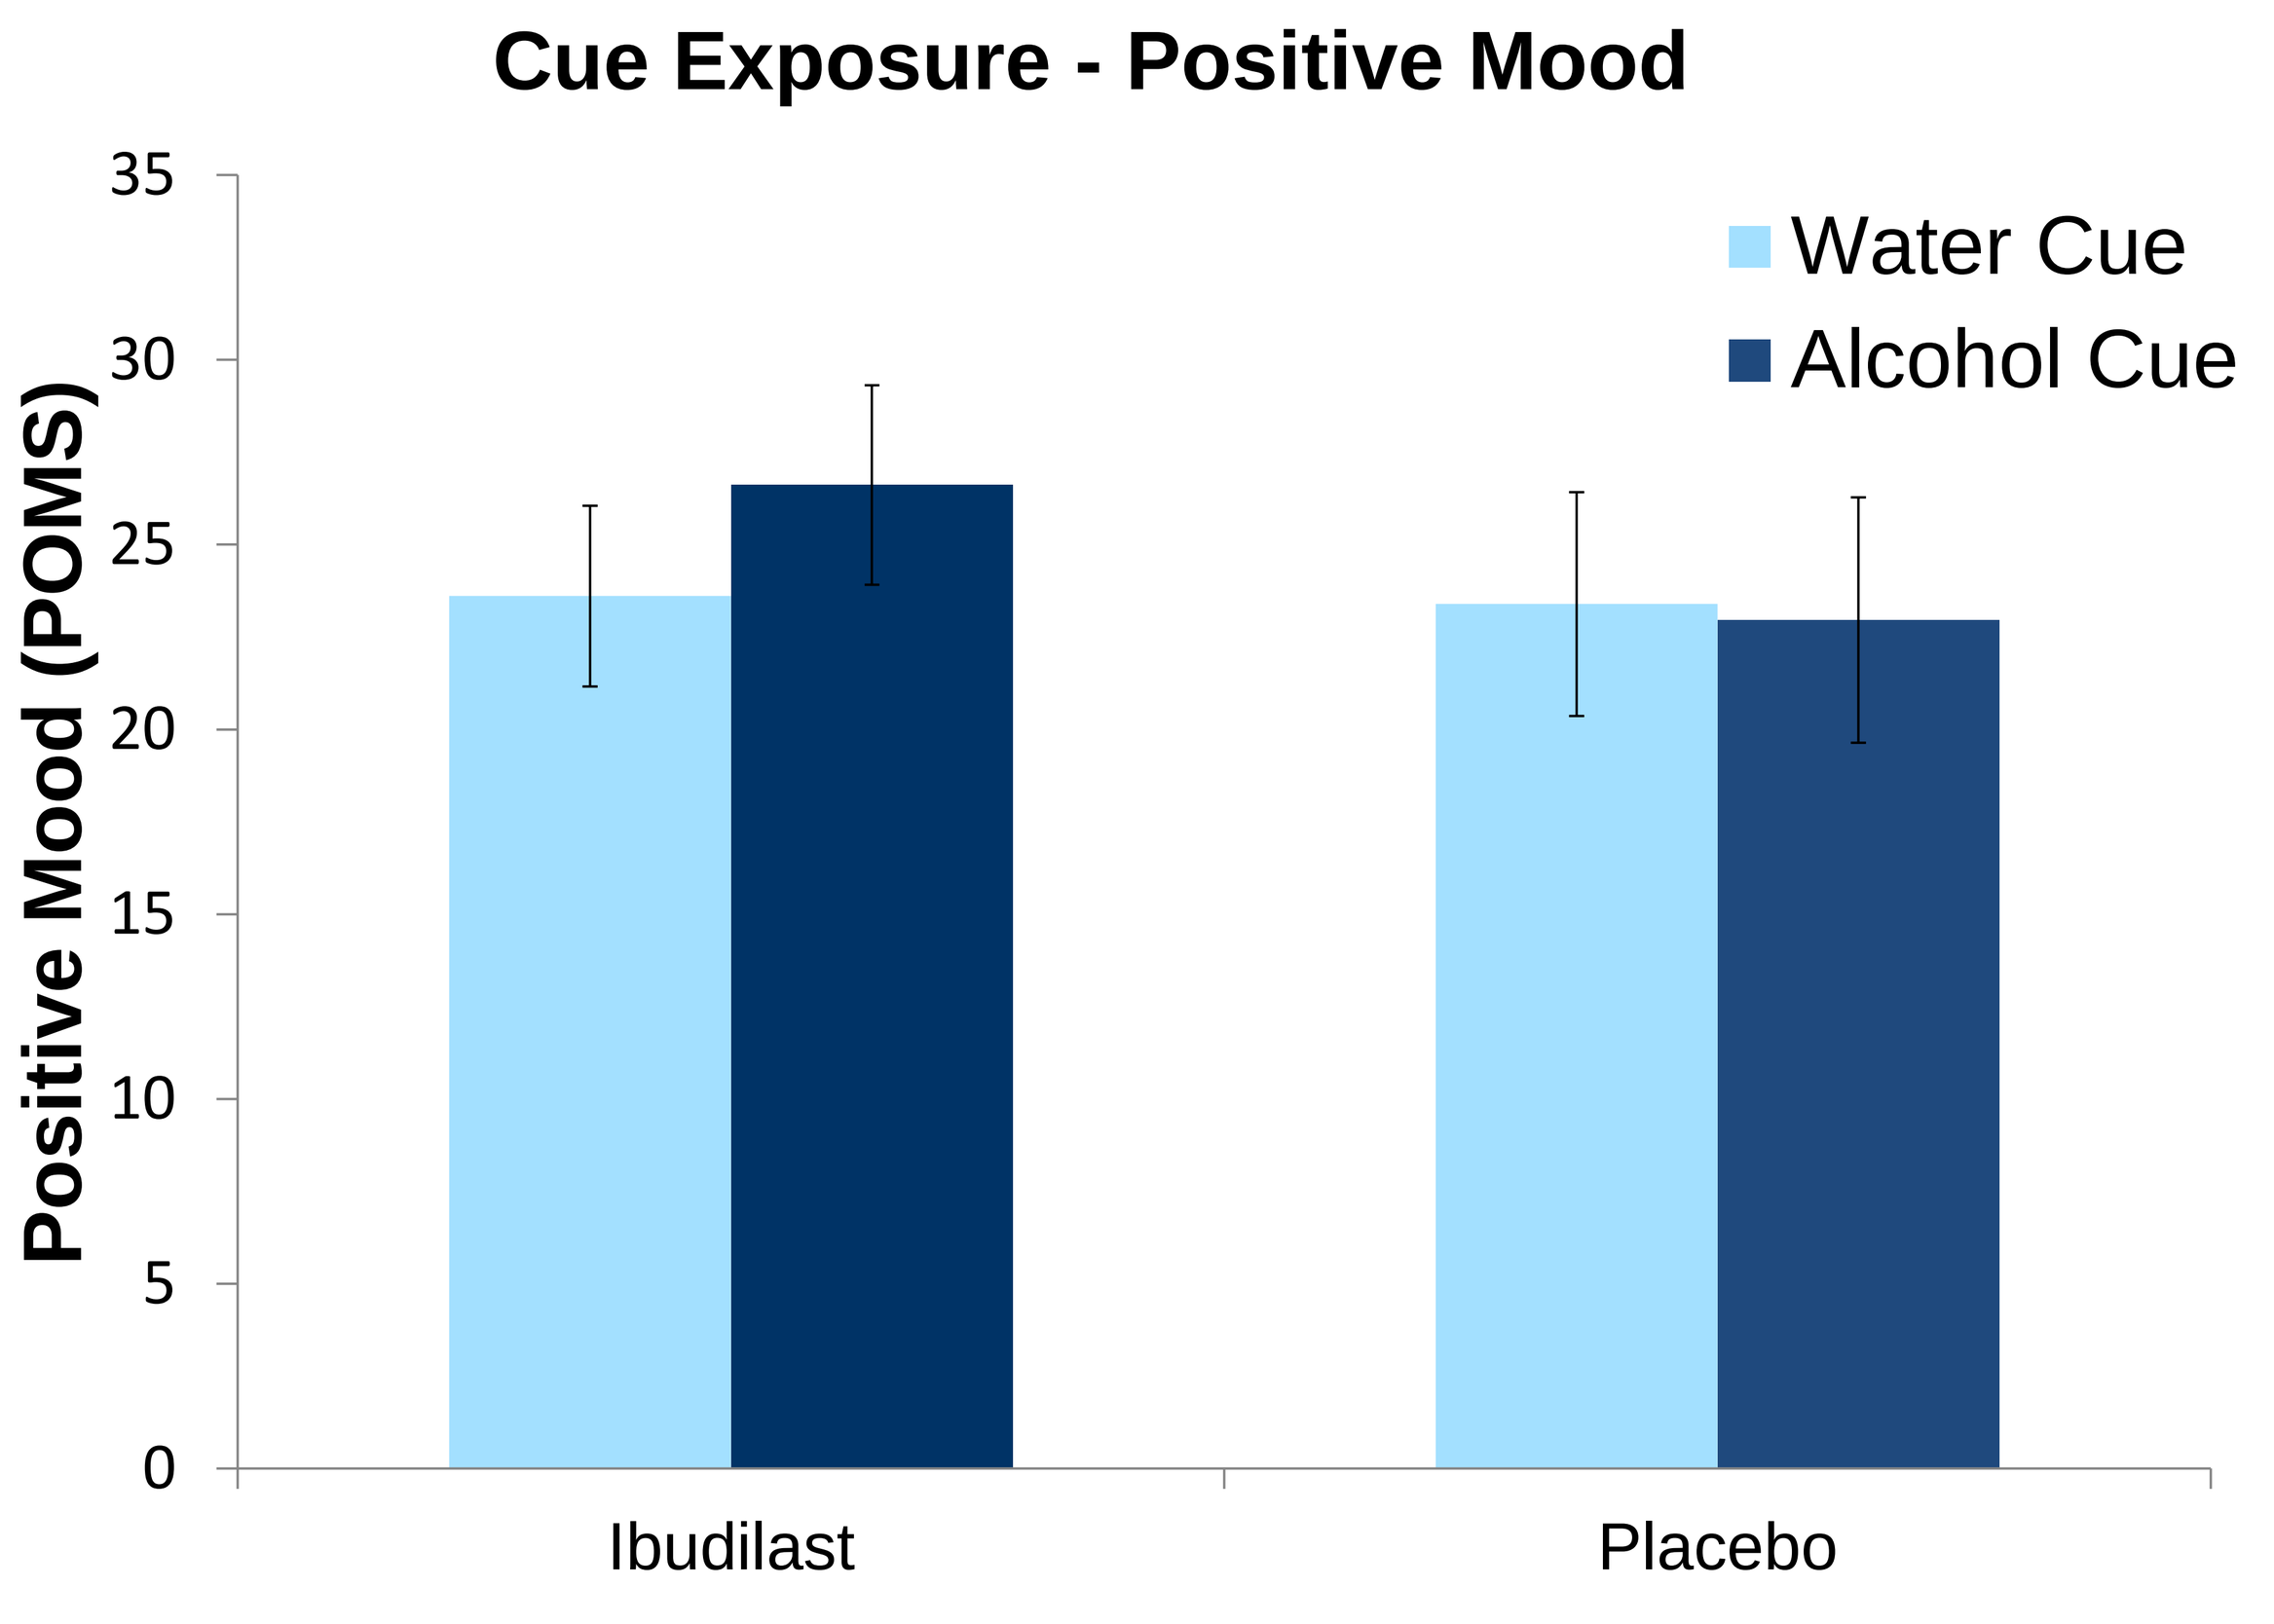
Cue Exposure - Positive Mood
Positive Mood (POMS)
0
5
10
15
20
25
30
35
Ibudilast
Placebo
Water Cue
Alcohol Cue

Medication × Cue, $F(1,22) = 3.62$, $p = .07$

Figure 3: Stress Reactivity

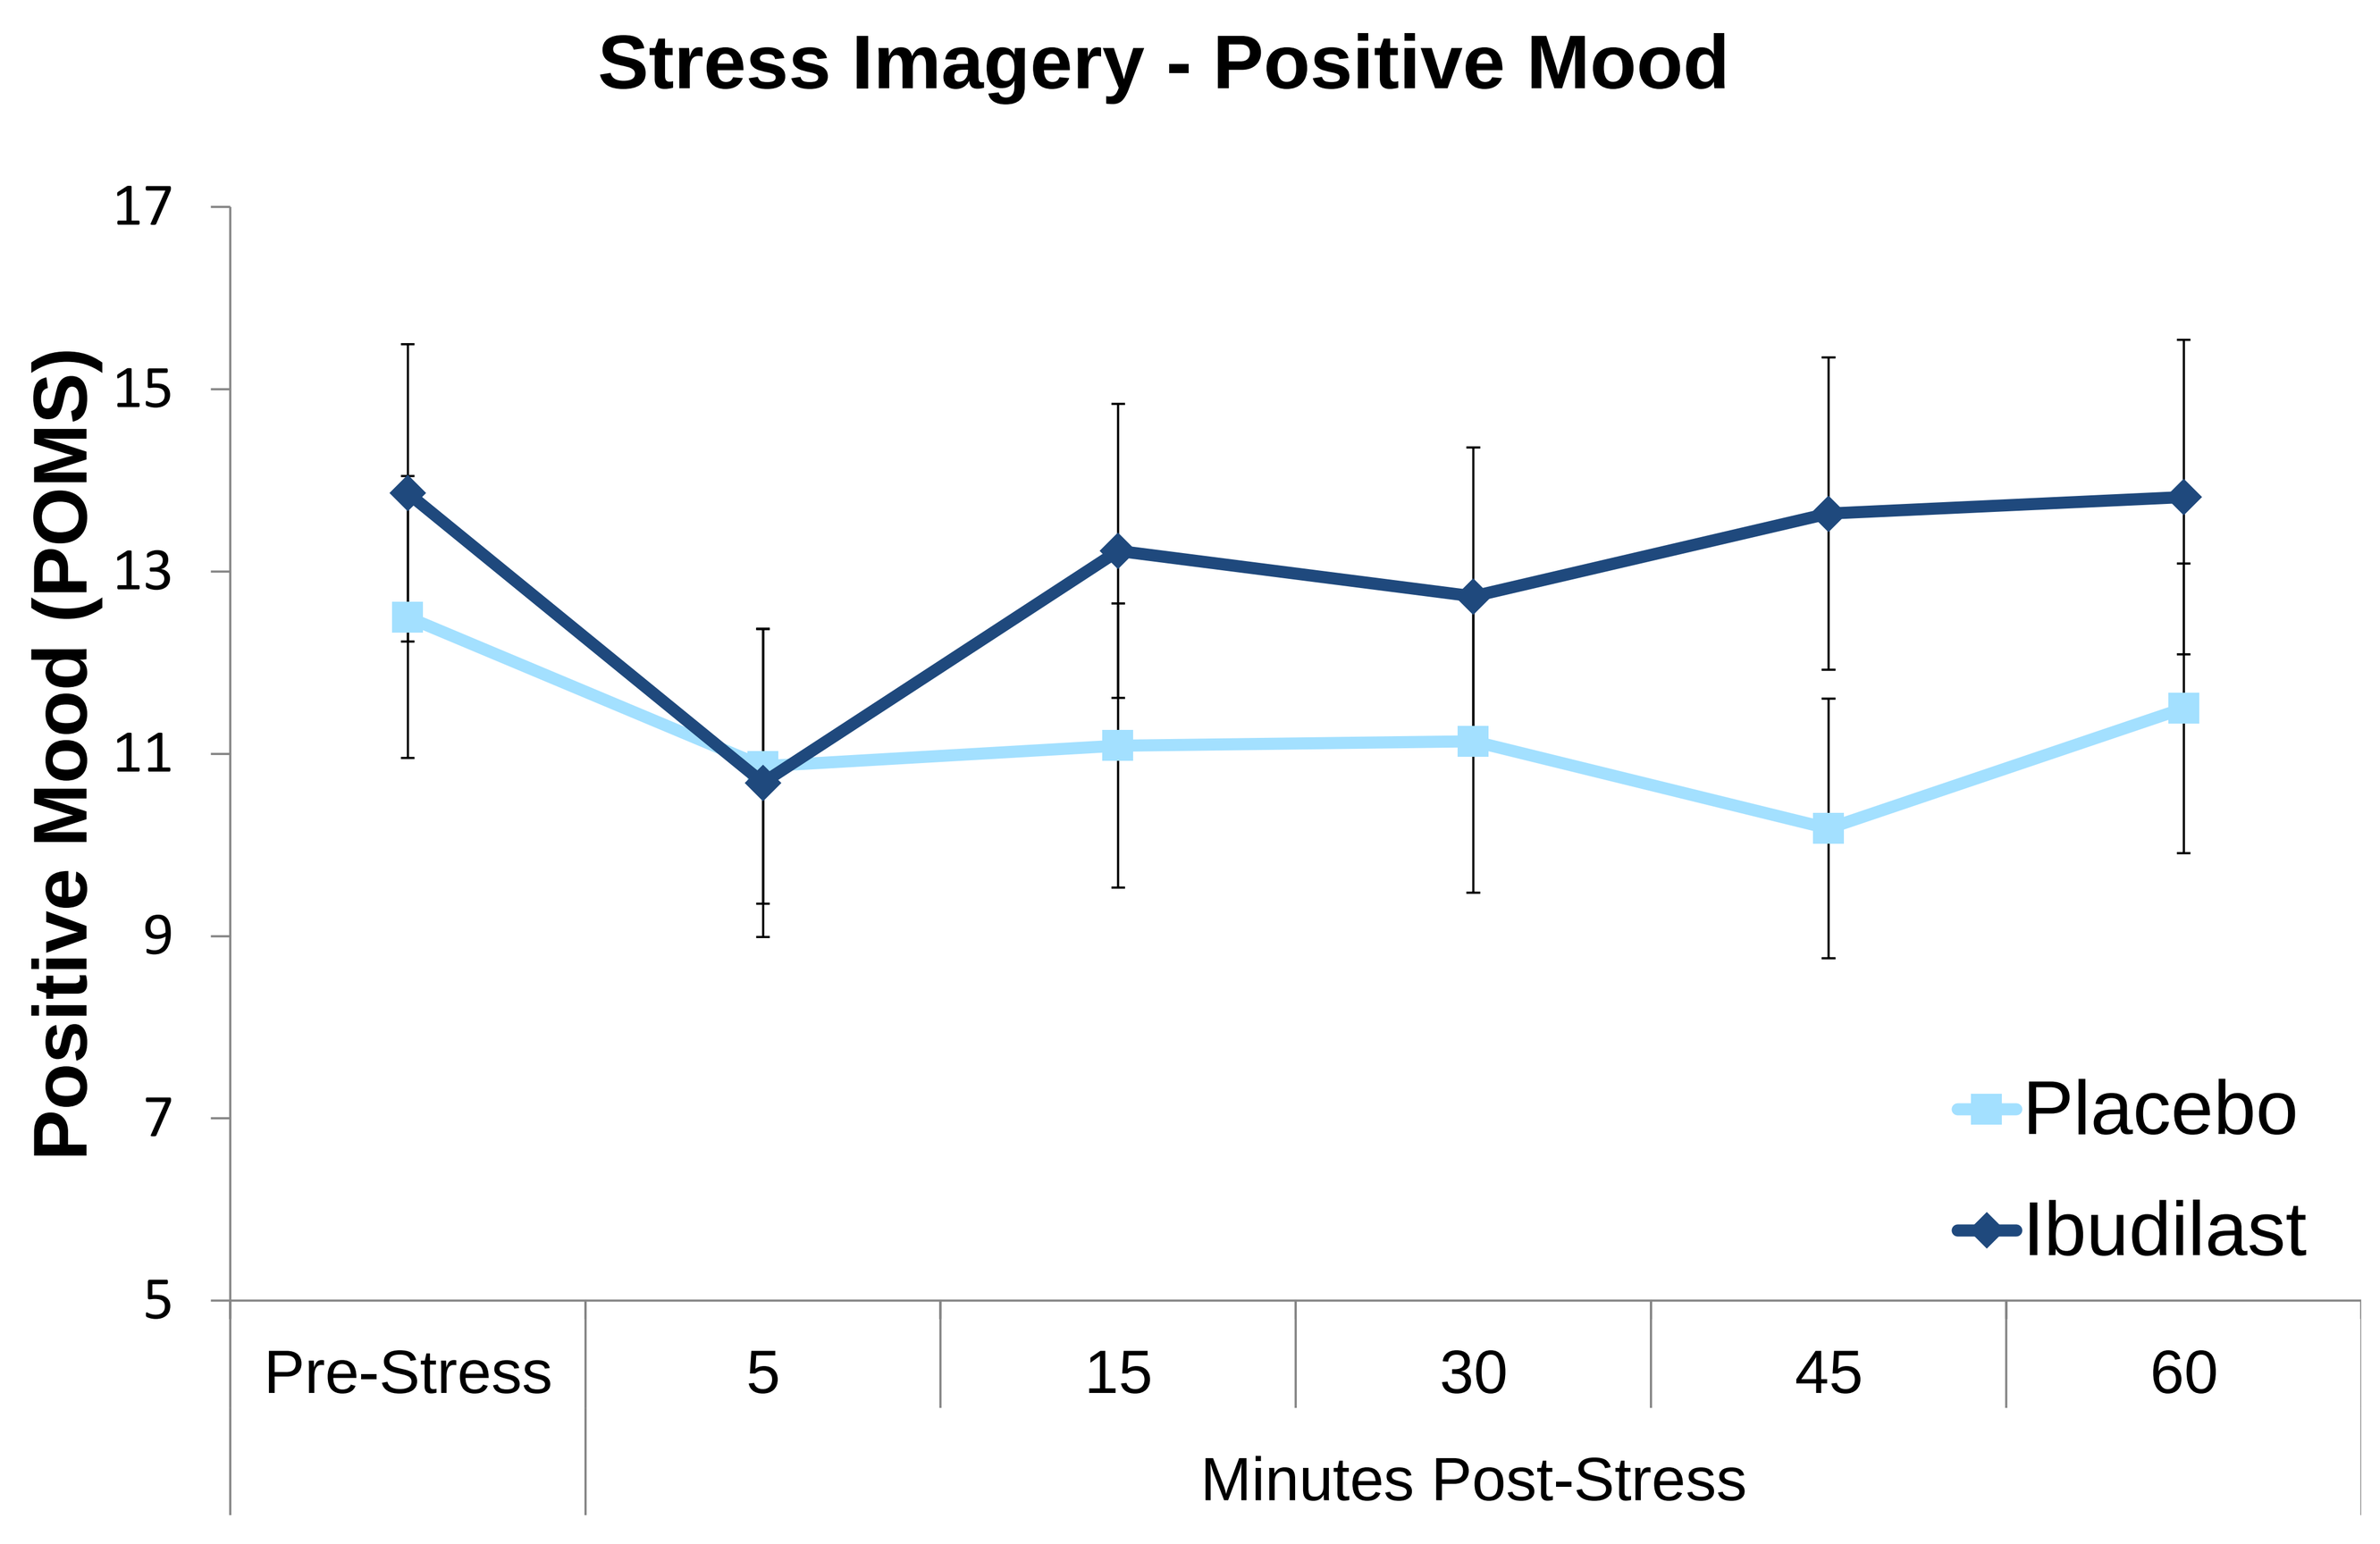
Stress Imagery - Positive Mood
Positive Mood (POMS)
5
7
9
11
13
15
17
Pre-Stress
5
15
30
45
60
Minutes Post-Stress
Placebo
Ibudilast

Medication × Time, $F(5,115) = 2.64$, $p < .05$

Figure 4: Cortisol Level

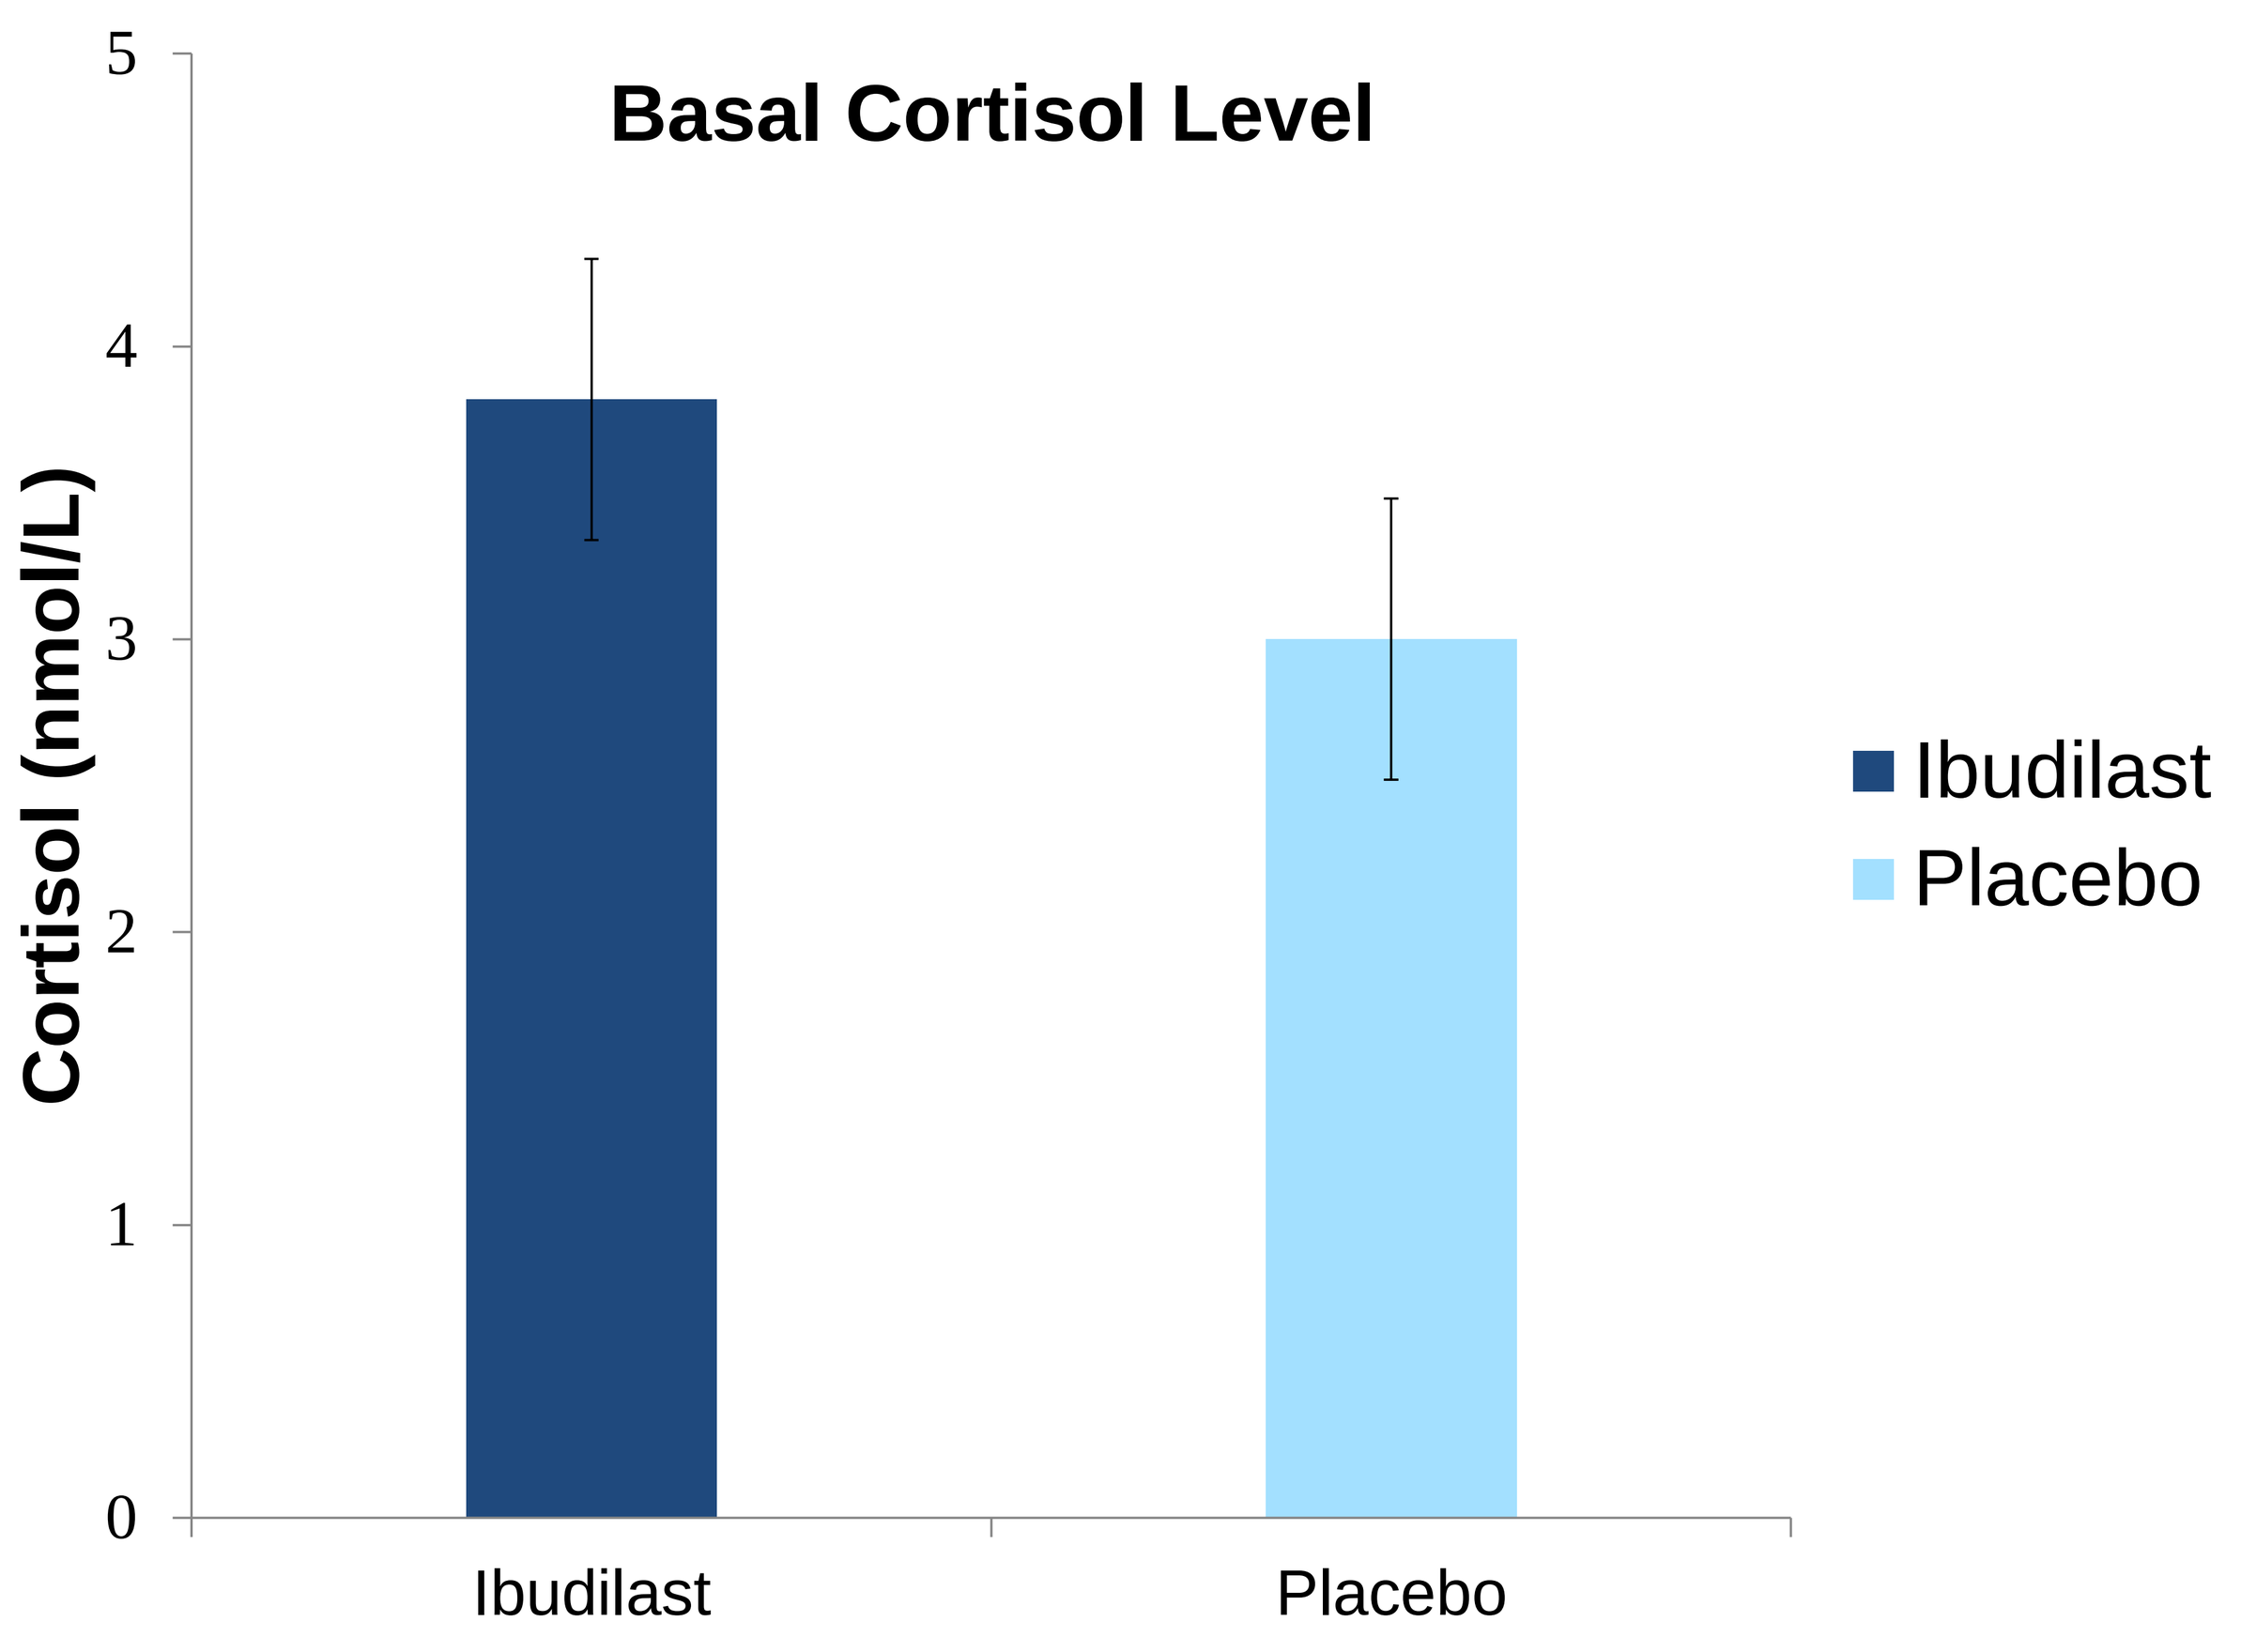
Basal Cortisol Level
Cortisol (nmol/L)
0
1
2
3
4
5
Ibudilast
Placebo
Ibudilast
Placebo

Medication, $F(1,21) = 3.53$, $p = .07$

CONCLUSIONS

Summary:

- The results of the present study indicate that IBUD (50 mg BID) may be a safe and promising treatment for AUD.
- IBUD reduced daily alcohol craving, produced a modest increase in basal cortisol levels, and promoted a sustained elevation in positive mood during exposure to alcohol-related cues and stressful imagery.

Implications & Future Directions:

- IBUD may be useful for treating AUD by enhancing positive mood, which could ameliorate the mood dysfunction often observed during protracted withdrawal.
 - These findings are also consistent with a hypothesized role for neuroinflammation in mood dysfunction.
- Additional studies of IBUD for AUD treatment appear warranted in larger studies, using chronic dosing and additional markers of efficacy.

Funding Source and Disclosures:

- This work and its authors were supported by the following grants: NIDA R21AA022214 (L.A.R.), NIAAA F31AA022569 (S.B.) TRDRP 23FT-0102 (D.J.O.R.). Lara A. Ray has received medication from Pfizer and Medicinova, a research award from Pfizer, and consulted for GSK. Medicinova, Inc. provided the study medication (IBUD).