Evaluation of MN-221 (bedoradrine), a novel, highly selective beta$_2$-adrenergic receptor agonist via intravenous infusion in mild to moderate asthma

Kazuko Matsuda, MD, Yuichi Iwaki, MD,
Maria Feldman and Alan Dunton, MD • MediciNova, Inc. San Diego, CA

1

Abstract

PURPOSE: MN-221 (bedoradrine) is a novel, highly selective beta$_2$-adrenergic receptor agonist under development for the treatment of acute exacerbation of asthma and chronic obstructive pulmonary disease (COPD). This study evaluated the safety and tolerability of MN-221 via intravenous infusion in patients with mild to moderate stable asthma.
METHODS: This was a randomized, double –blind, placebo controlled, multi-center sequential dose escalation study. Eligible patients were enrolled and randomized to one of the four dose-escalation sequences. Adverse events (AEs) and other clinical safety measures were recorded.
RESULTS: Seven dose levels were evaluated. Doses below 150ug were safe but did not produce clinically significant changes in FEV$_1$. We report here doses of 150ug to 900ug. A total of 23 patients were enrolled. An overall dose response in mean change in FEV$_1$ from pre-infusion to end of infusion with MN-221 was observed. Mean changes of FEV$_1$ at 150µg, 240µg, 450µg and 900µg of MN-221 were 8.0%, 7.2%, 11.6% and 8.1% respectively and those changes were all statistically significant compared to the placebo ranging from p<0.001 to p<0.0001. No serious or severe AEs, or discontinuation due to AEs were reported. Most clinical laboratory results were within normal ranges or not clinically significant. Dose related heart rate increase was observed but not considered clinically significant because symptoms associated with heart rate increase were not observed.
CONCLUSION: MN-221 was safe and well tolerated in mild to moderate stable asthma patients. Preliminary evidence of MN-221 efficacy was observed as dose dependent improvements in FEV$_1$.
CLINICAL IMPLICATIONS: MN-221 may represent a safe and novel adjunctive approach for treating acute asthma exacerbations.

2

MN-221 (bedoradrine sulfate)

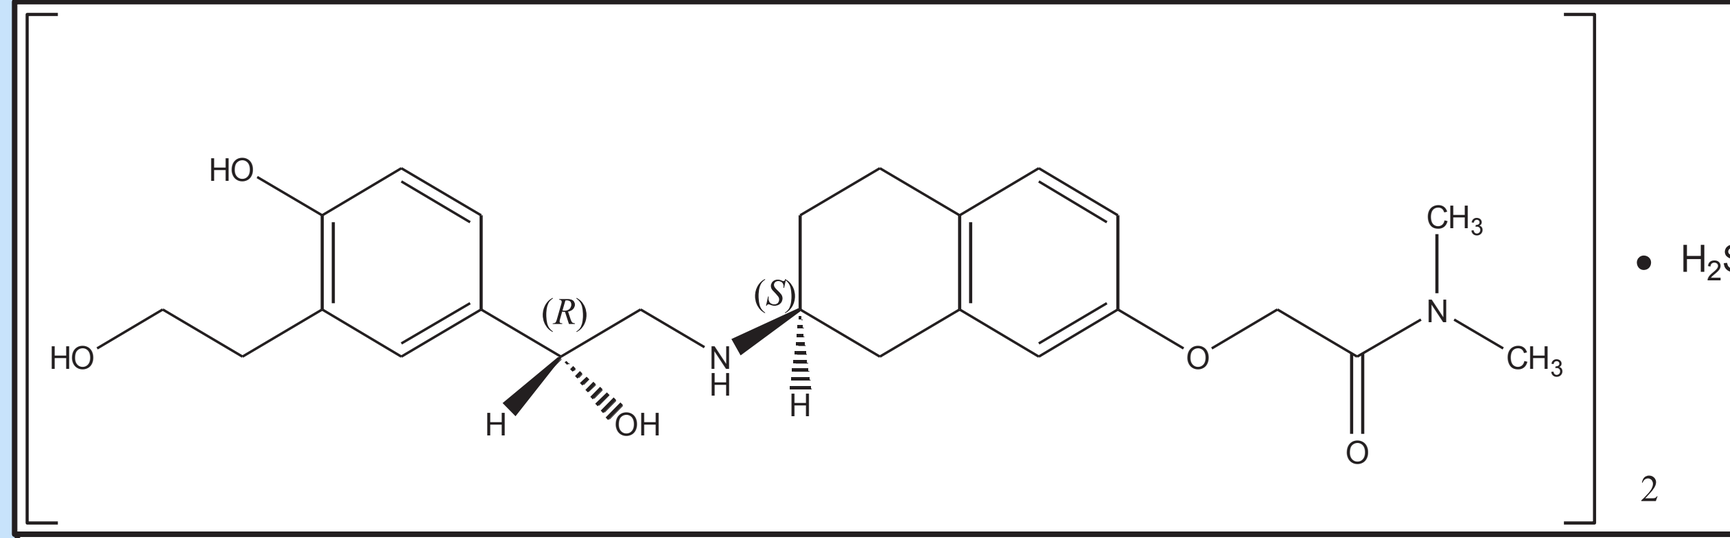
HO
HO
(R)
H
OH
N
H
(S)
H
O
O
CH3
N
CH3
2
• H2SO4

- **High beta$_2$ Potency: EC$_{50}$[a] ~ 3 nM**
- **High beta$_2$ selectivity[a]: beta$_2$/beta$_1$ > 800**
 - **≥ 4 fold greater beta$_2$ vs. beta$_1$ selectivity when compared alongside Terbutaline, Albuterol, Levalbuterol**
 - **beta vs. alpha adrenoceptor (or other receptor systems) > 250**
- **Partial beta$_1$ agonist, full beta$_2$**
- **in vivo duration of action > SABA, <LABA**

[a] Inoue et al., J. Obst. Gynec. Res. 35:405, 2009

3

MN-221: More selective than other beta-agonists

Test Drug	beta$_1$IC$_{50}$ (M)	beta$_2$IC$_{50}$ (M)	Beta$_2$-adrenoceptor Selectivity (IC$_{50}$ for beta$_1$ / IC$_{50}$ for beta$_2$)
Levalbuterol	7.40E-06	1.40E-06	5.3
Albuterol	9.40E-06	1.60E-06	5.9
Terbutaline	6.00E-05	6.50E-06	9.2
MN-221	5.90E-06	1.40E-07	42.4

4

Purpose

- Determine safety and efficacy of i.v. MN-221 in subjects with mild to moderate asthma.
- Determine the pharmacokinetic (PK) profile.
- Dose guiding for Phase 2 trial(s) in acute exacerbation of asthma patients.

5

Study Design

- Randomized, placebo-controlled, double-blind, dose escalation study
- 23 subjects with mild-to-moderate stable asthma (FEV$_1$ ≥ 60% predicted) at 4 sites. 15 minute i.v. infusions.
- Patients were randomized to one of four different treatment groups (3:1 active:placebo)
 - The first dose was 0.35 µg/min for 15 minutes, subsequent doses were 1.0 µg/min, 3.5 µg/min, 10 µg/min, 16.0 µg/min, 30.0 µg/min, and 60.0 µg/min.
 - Each treatment sequence consisted of placebo and escalating doses of MN-221 (total doses of 5.25 µg, 15.0 µg, 52.5 µg, 150 µg, 240 µg, 450 µg, 900 µg)

6

Dose-Escalating Treatment Sequences (15 min i.v. infusions)

	Group A	Group B	Group C	Group D
Visit 2	Placebo	0.35 µg/min	0.35 µg/min	0.35 µg/min
Visit 3	1.0 µg/min	Placebo	1.0 µg/min	1.0 µg/min
Visit 4	3.5 µg/min	3.5 µg/min	Placebo	3.5 µg/min
Visit 5	10 µg/min	10 µg/min	10 µg/min	Placebo
Visit 6	Placebo	16 µg/min	16 µg/min	16 µg/min
Visit 7	30 µg/min	Placebo	30 µg/min	30 µg/min
Visit 8	60 µg/min	60 µg/min	Placebo	60 µg/min

7

Efficacy & Safety Endpoints

- Primary efficacy endpoint was the change from pre-infusion to post-infusion FEV$_1$
- Secondary efficacy endpoint was the change in FEV$_1$ at all post-infusion timepoints and the change in PEFR at each scheduled post-dose timepoint
- Safety was evaluated by adverse events (AEs), clinical laboratory findings, physical examinations findings, electrocardiograms (ECGs), and changes in vital signs

8

Major Inclusion/Exclusion Criteria

Subjects meeting all of the following were considered for admission to the study:

- Male or female 18 to 50 y.o.;
- Asthma ≥ 3 months with a pre-bronchodilator FEV$_1$ ≥ 60% of predicted, and not receiving inhaled corticosteroids;
- An increase in FEV$_1$ of at least 12%, and of at least 200 cc, over the pre-albuterol FEV$_1$ within 30 minutes after inhalation of up to 4 puffs of albuterol via metered dose inhaler at the Screen Visit.

Subjects were excluded from the study if they met any of the following criteria:

- Emergency treatment for asthma within 1 month, or hospitalized for asthma within 3 months of Screen Visit 1;
- An upper or lower respiratory tract infection within 3 weeks, or sinus infection within 7 days of Screen Visit 1;
- Had taken any of the excluded asthma/allergy medications.

9

MN-221 Mean Change in FEV$_1$ (% Predicted) at End of Infusion

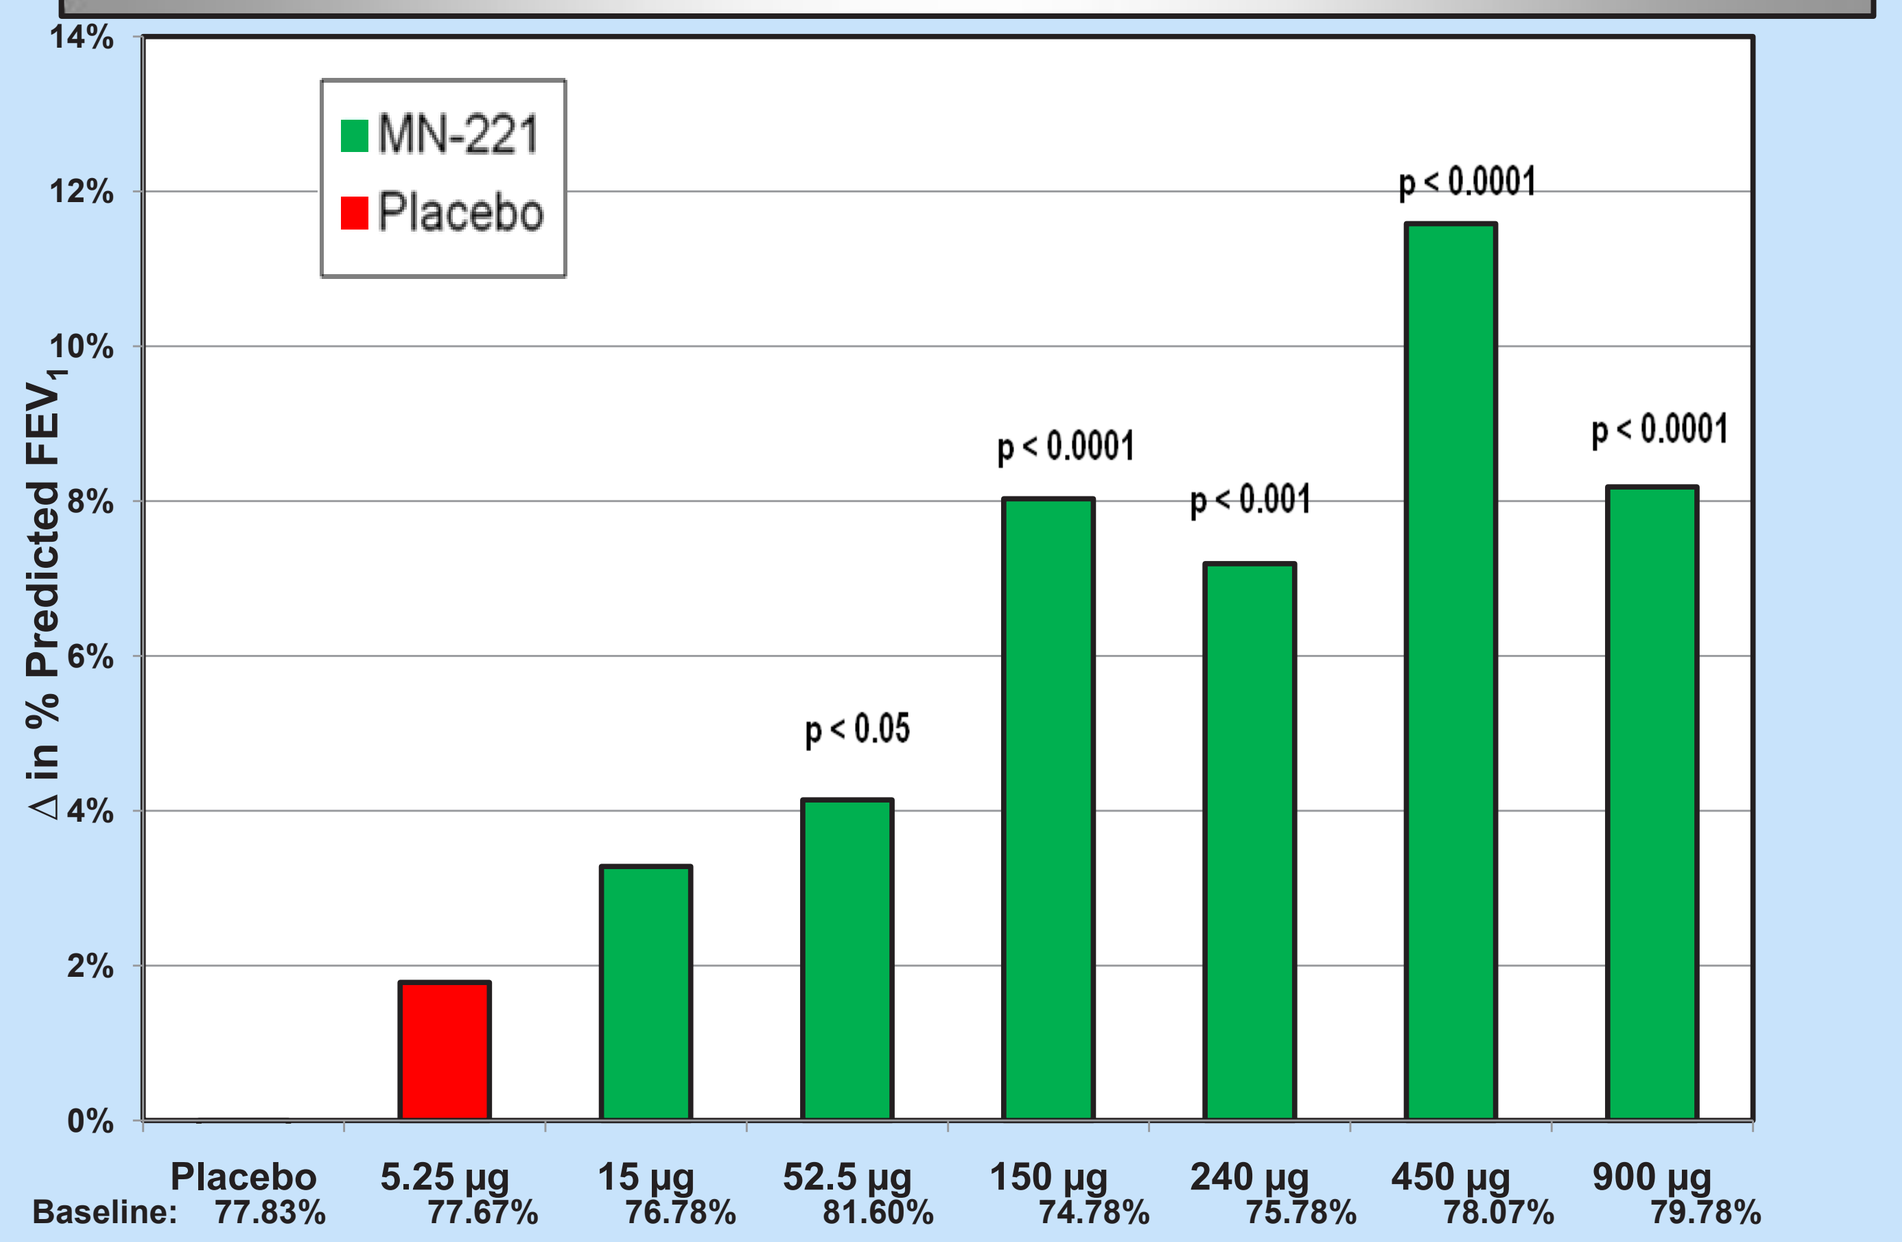
MN-221
Placebo
14%
12%
10%
8%
6%
4%
2%
0%
Δ in % Predicted FEV1
p < 0.05
p < 0.0001
p < 0.001
p < 0.0001
p < 0.0001
Placebo
5.25 µg
15 µg
52.5 µg
150 µg
240 µg
450 µg
900 µg
Baseline: 77.83% 77.67% 76.78% 81.60% 74.78% 75.78% 78.07% 79.78%

10

Analysis of Changes in Percent Predicted FEV$_1$ At 1 Hour

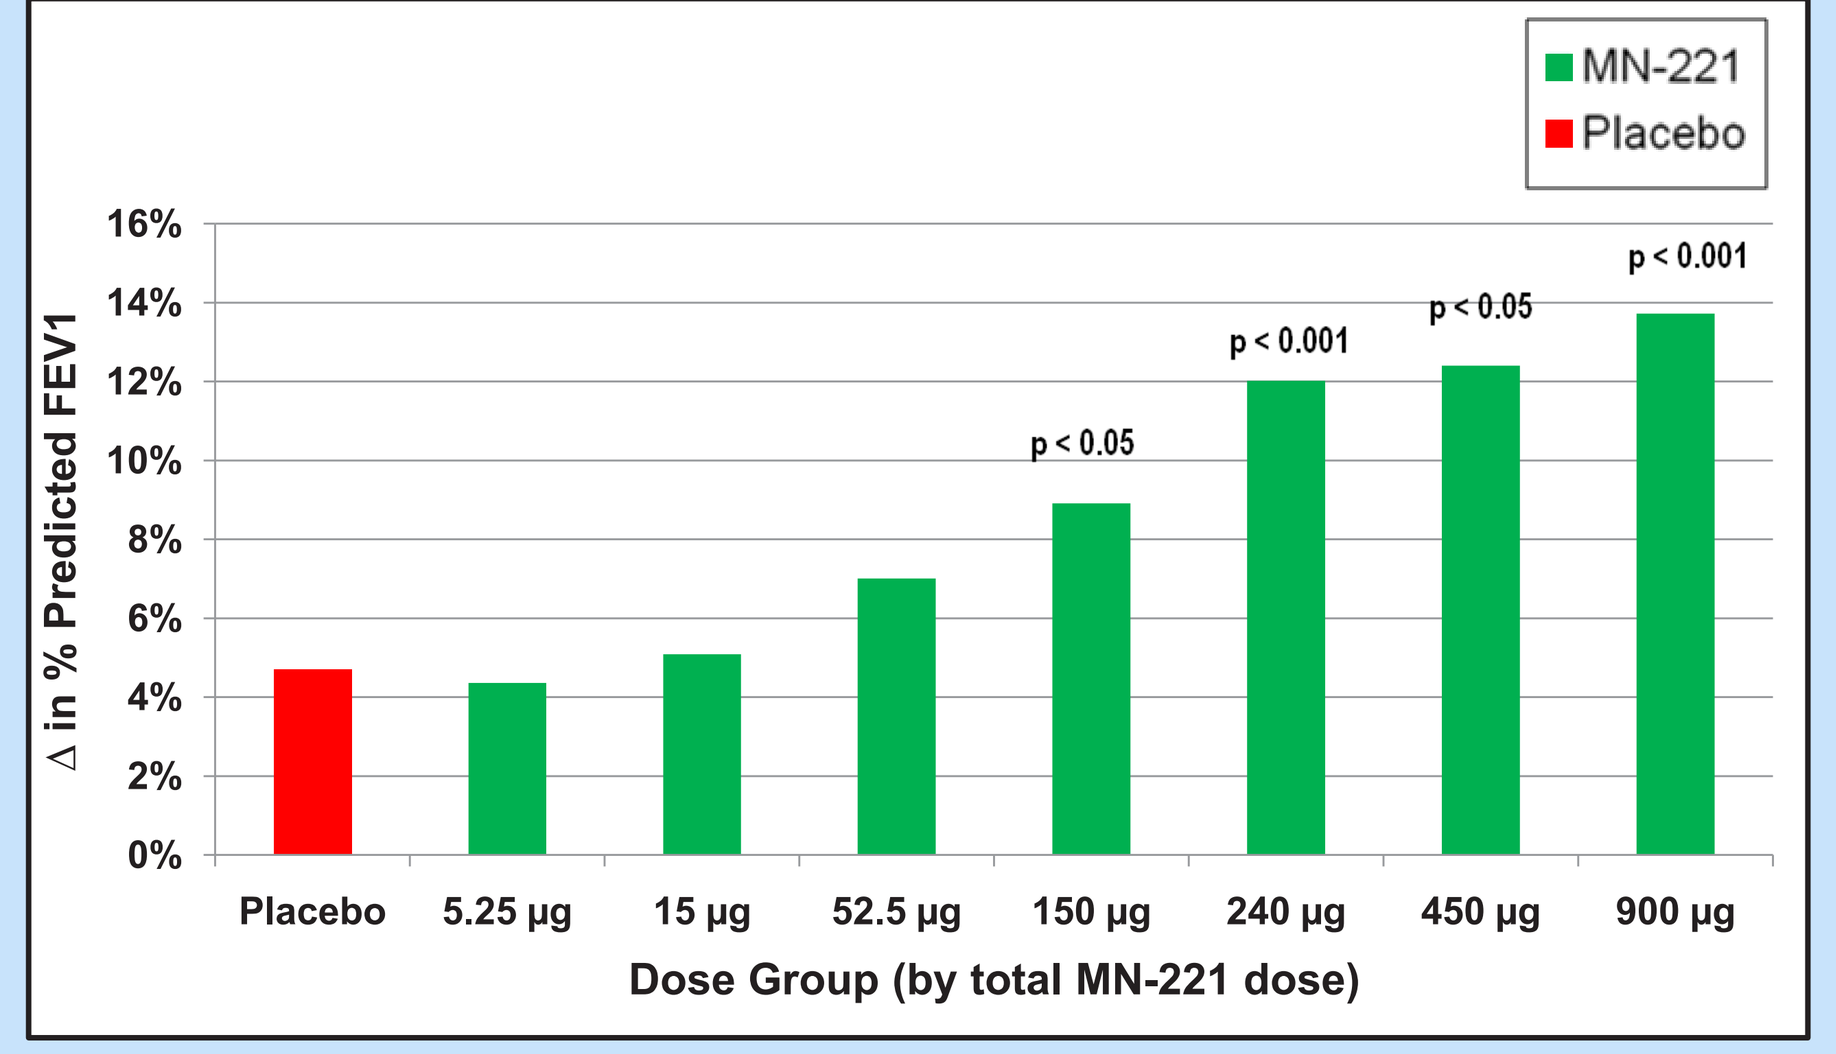
MN-221
Placebo
16%
14%
12%
10%
8%
6%
4%
2%
0%
Δ in % Predicted FEV1
p < 0.05
p < 0.001
p < 0.05
p < 0.001
Placebo
5.25 µg
15 µg
52.5 µg
150 µg
240 µg
450 µg
900 µg
Dose Group (by total MN-221 dose)

11

Analysis of Changes in Heart Rate (bpm) from Start of Infusion

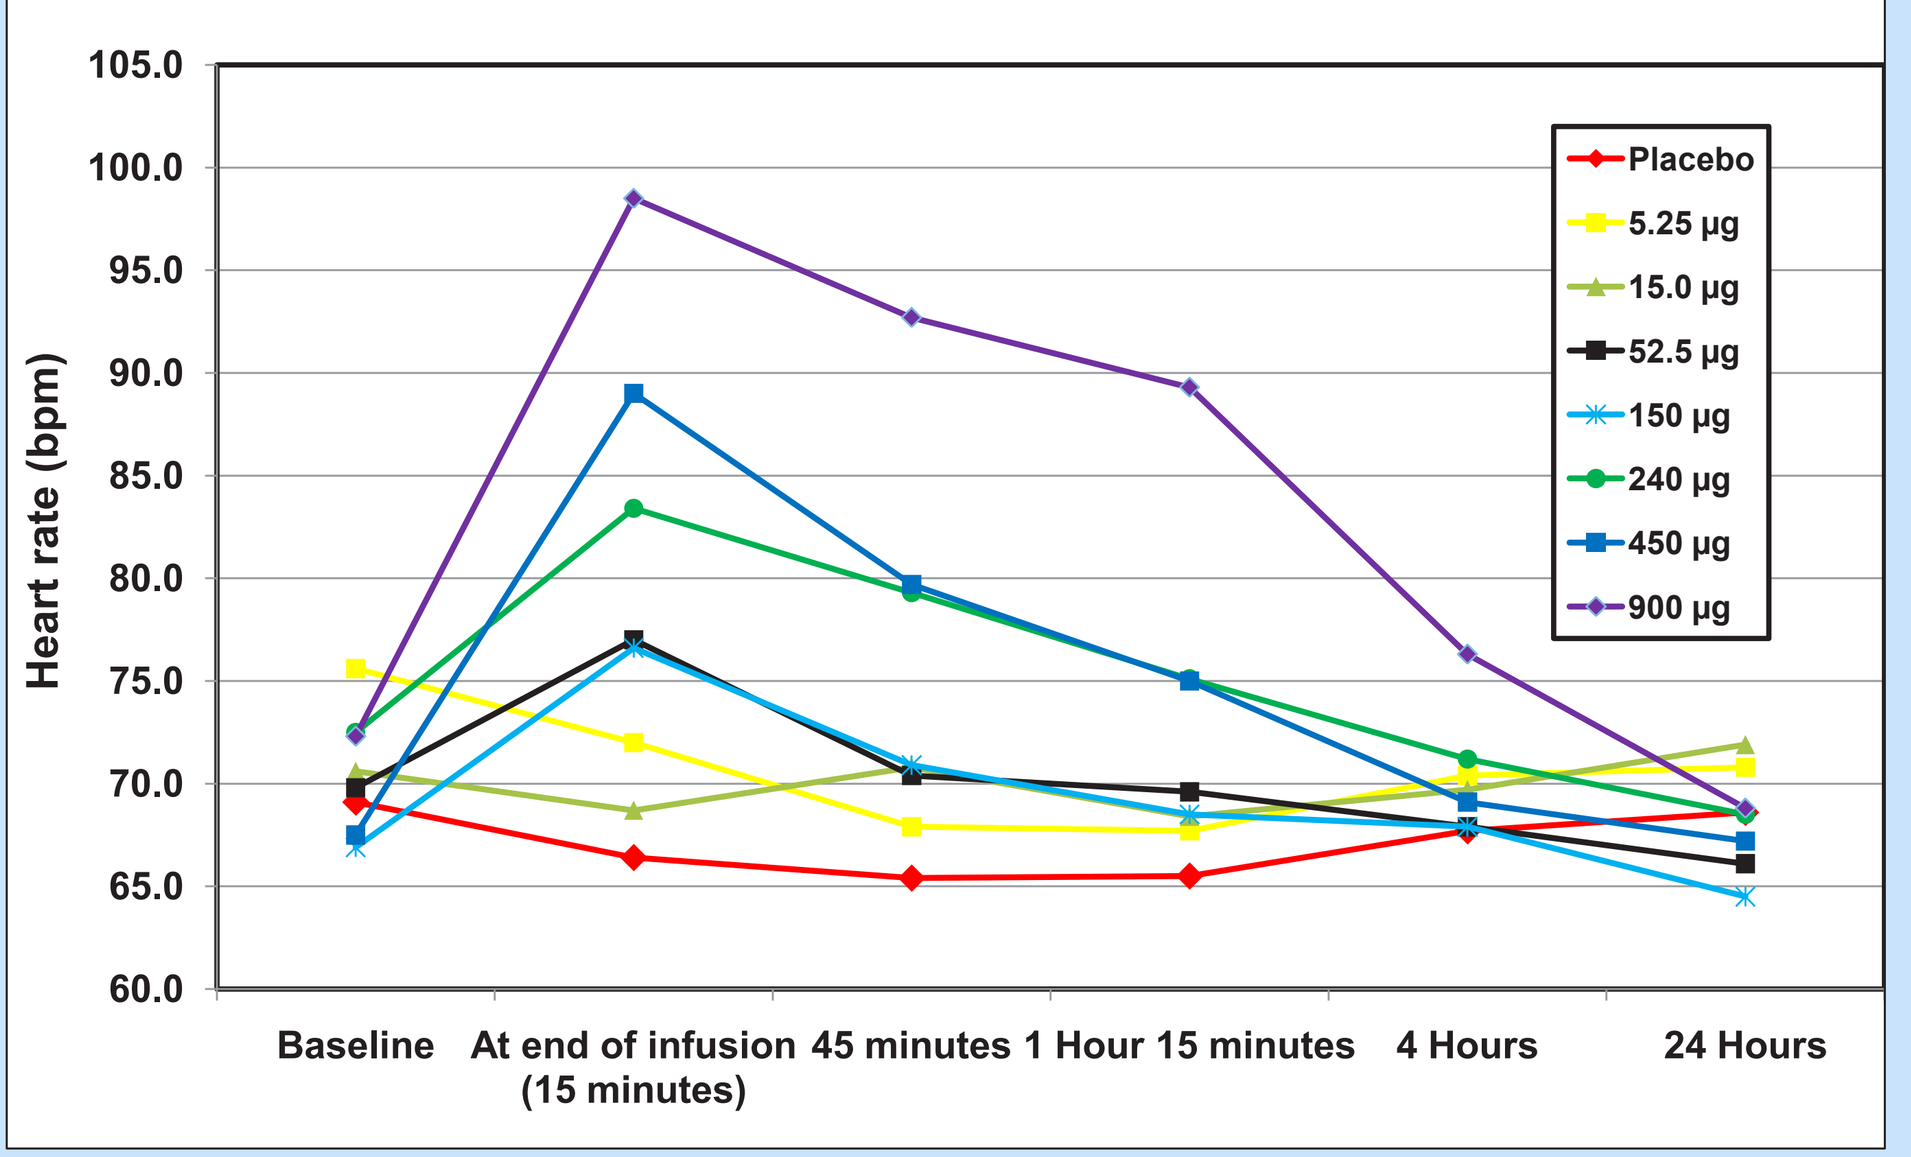
105.0
100.0
95.0
90.0
85.0
80.0
75.0
70.0
65.0
60.0
Heart rate (bpm)
Placebo
5.25 µg
15.0 µg
52.5 µg
150 µg
240 µg
450 µg
900 µg
Baseline
At end of infusion (15 minutes)
45 minutes
1 Hour 15 minutes
4 Hours
24 Hours

12

MN-221 Safety & PK Conclusions

- Infusions of all doses of MN-221 were well tolerated in this patient population.
 - Maximum heart rates remained below 100 bpm
 - No adverse QT changes
 - No SAEs or dropouts from study due to AE
- PK of MN-221 was linear and dose proportional from 0.35 through 60 µg/min MN-221
- Elimination half-life was ~10 hr and consistent across doses

13

MN-221 Efficacy Summary

- i.v. infusions of MN-221 were efficacious in patients with mild to moderate asthma
 - Significant increases in FEV$_1$(L) and FEV$_1$ (%Pred) vs placebo
 - Dose response in FEV$_1$ increases at each time point through 1 hr post-infusion in the ITT population; at 4 and 8 hr for the protocol-correct population
 - Optimal effect at 450 and 900 µg.
- Mean changes in PEFR were statistically significant in the 450 and 900 µg doses of MN-221 versus placebo.

14

Clinical Implications

- MN-221 appeared to be safe and well-tolerated in stable mild to moderate asthma patients
- Preliminary evidence of MN-221 efficacy was observed with dose-dependent improvements in FEV$_1$
- MN-221 (bedoradrine) may represent a safe and novel adjunctive approach for treating acute asthma exacerbations