Reduced Hospital Admission and Improved Pulmonary Function Following Intravenous MN-221 (Bedoradrine), a Novel Highly Selective Beta$_2$-Adrenergic Receptor Agonist, Adjunctive to Standard of Care in Severe Acute Exacerbation of Asthma

Richard Nowak, MD[2], Yuichi Iwaki, MD[1], Kazuko Matsuda, MD[1], Kirk Johnson, PhD[1], and Alan Dunton, MD[1]
[1]MediciNova Inc., San Diego, CA • [2]Henry Ford Health System, Detroit, MI

1

Abstract

PURPOSE: Acute exacerbations of asthma are an increasingly important and costly burden. This study was designed to assess the safety and efficacy of intravenous MN-221 as add-on to standard of care (SOC) in an Emergency Department (ED) setting.
METHODS: The study was randomized, placebo-controlled, dose-escalation, and multicenter. 29 patients received SOC (nebulized albuterol and ipratropium plus oral corticosteroid). Patients with FEV$_1$ ≤ 55% of predicted were randomized to MN-221 [240 or 450 mg infused over 15 min or 1080 mg over 2 hr] or Placebo. Safety, efficacy and PK parameters were monitored hourly throughout a 5 hr treatment period and a 24 hr follow-up visit. Nebulized albuterol (2.5 mg) and/or ipratropium (0.5 mg) was available hourly during screening and treatment phases. Efficacy endpoints included spirometry, dyspnea indexing, albuterol use, and hospitalization rates.
RESULTS: The study was completed with n=13 in SOC + Placebo and n=16 receiving SOC + MN-221; 5 at 240 mg, 6 at 450 mg, and 5 intended for a 1080 mg dose (2 patients actually received 1995 mg). MN-221 was well-tolerated. MN-221 efficacy included: 1) reduced hospitalization rate: 4/16 (25%) in the all MN-221 group $_1$ 7/13 (54%) in Placebo arm, and 2) improved FEV$_1$ (% predicted): it was elevated in the all MN-221 group (change from baseline in AUC1-5hr was 43% higher in the all MN-221 group vs. Placebo). The majority of adverse events were mild to moderate in intensity and SAEs were lower in the all MN-221 group vs. Placebo and were asthma AEs. No abnormal ECG findings were observed and heart rate was minimally elevated in the all MN-221 group relative to Placebo.
CONCLUSIONS: MN-221 adjunctive to standard therapy for severe acute asthma exacerbations was safe and appeared to provide additional clinical benefit. The results support further clinical development of MN-221.
CLINICAL IMPLICATIONS: MN-221 appears to be a unique, safe, and clinically-beneficial adjunctive pharmacotherapy for the treatment of acute asthma. Further development is warranted.

2

Exacerbation of Asthma

- An exacerbation of asthma is an acute episode of progressively worsening shortness of breath, cough, wheezing, and chest tightness, or some combination of these symptoms that are characterized by decreases in expiratory airflow
- Exacerbations can vary in severity from mild to life-threatening and can occur in patients with any level of asthma (i.e., intermittent, or mild, moderate, or severe persistent asthma)
- In severe episodes, bronchial spasm, airway inflammation, and mucous plugging leading to progressive increases in airflow resistance, result in difficulty breathing, carbon dioxide retention, hypoxemia, respiratory muscle fatigue, and respiratory failure

3

Conventional Pharmacotherapy for Acute Exacerbation of Asthma

- Beta agonist agents are the mainstays of acute therapy in asthma
 - They act as bronchodilators to relax bronchial smooth muscle, increase mucociliary clearance, decrease mucous production, and may inhibit the release of mast cell mediators
 - The inhaled route of administration is first line route of delivery for most patients
 - Parenteral therapy may be given by subcutaneous (SC) or intravenous (IV) administration, or, in an emergency, via endotracheal tube
- Isoproterenol, terbutaline, and albuterol cause potent beta$_1$ stimulation which may lead to significant tachycardia, hypokalemia, dysrhythmias, tremor, and inotropy
- Corticosteroids, e.g., methylprednisolone or prednisone, work largely on the late phase reaction in asthma, causing a delay in onset of action

4

MN-221 (bedoradrine sulfate)

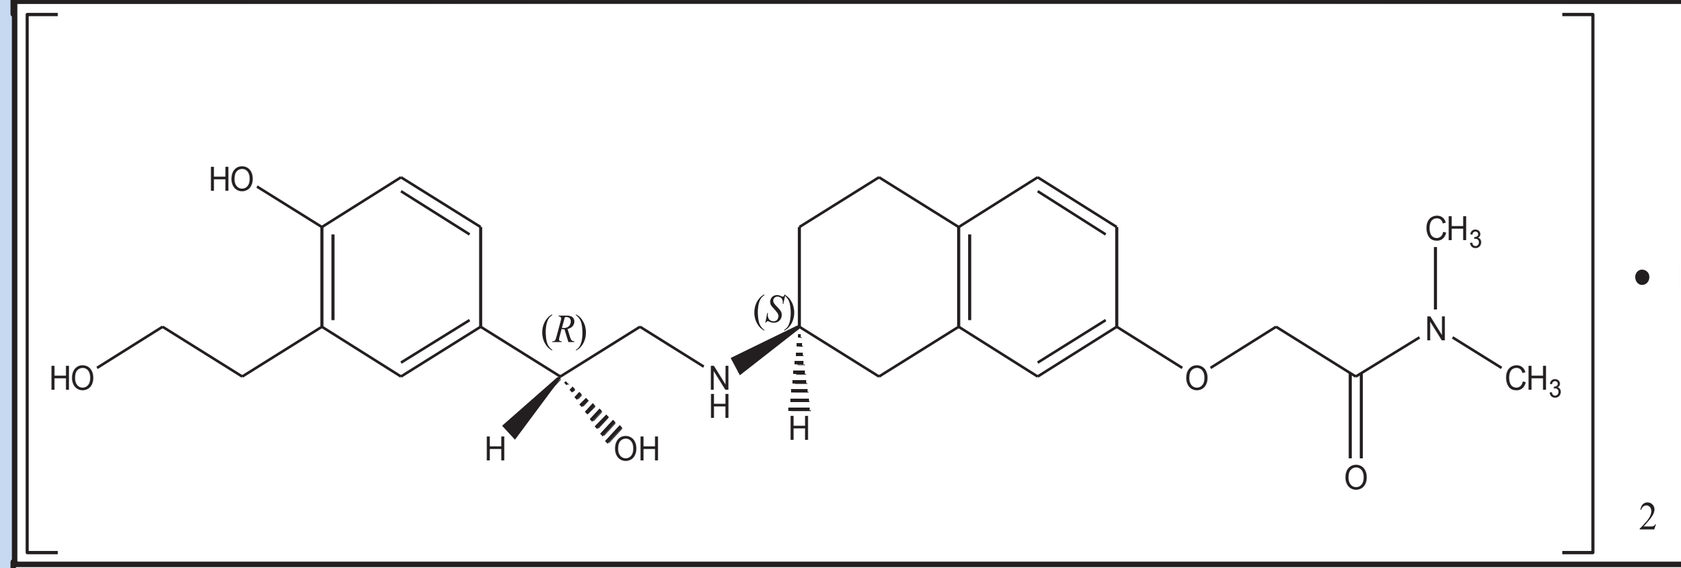
HO
HO
(R)
(S)
N
H
OH
CH$_3$
N
CH$_3$
O
O
• H$_2$SO$_4$
2

- **High beta$_2$ Potency: EC$_{50}$[a] ~ 3 nM**
- **High beta$_2$ selectivity[a]: beta$_2$/beta$_1$ > 800**
 - **≥ 4 fold greater beta$_2$ vs. beta$_1$ selectivity when compared alongside Terbutaline, Albuterol, Levalbuterol**
- **beta vs. alpha adrenoceptor (or other receptor systems) > 250**
- **Partial beta$_1$ agonist, full beta$_2$**
- **In vivo duration of action > SABA, <LABA**

[a] Inoue et al., J. Obst. Gynec. Res. 35:405, 2009

5

Purpose

- To assess the safety and efficacy of intravenous (IV) MN-221 to patients experiencing exacerbation of asthma as add-on to standard of care (SOC) in an Emergency Department (ED) setting
- Safety assessments included (AEs), labs, pulse oximetry, physical exams, 12-lead ECG and Holter monitoring, vital signs, and concomitant medications (CMs)
- Pharmacokinetic (PK) assessment of MN-221 and R- and S-albuterol.
 - Results for comparison of MN-221 after IV administration in patients with acute asthma and those in subjects in prior studies with mild to moderate asthma
- Albuterol and MN-221 data used for PK and pharmocodynamic (PD) analyses

6

Major Inclusion Criteria

Subjects meeting all of the following were considered for admission to the study:
- Male or female 18 to 55 y.o.
- Asthma ≥ 3 months
- Diagnosis of an acute exacerbation of asthma upon presentation at the ED
- Upon presentation to the ED the treatment provided included:
 - Medical history and physical exam including vital signs, auscultation, assessments of accessory respiratory muscle usage and level of dyspnea
 - Oxygen given to maintain oxygen saturation measured by pulse oximetry of ≥ 90%
 - 2 doses inhaled beta$_2$-agonist (albuterol 5 mg) via nebulizer (each dose given sequentially up to approximately every 20 minutes), ***simultaneously with***
 - 2 doses of inhaled anti-cholinergic agent (ipratropium 0.5 mg) via nebulizer (each dose given sequentially up to approximately every 20 minutes)
 - 1 dose of corticosteroid of at least 60 mg given orally (prednisone) or intravenously (methylprednisolone)
- FEV$_1$ ≤ 55% within 10 minutes of completing the treatment described in the previous Inclusion Criterion

7

Major Exclusion Criteria

Subjects were excluded from the study if they met any of the following criteria:
- COPD or other chronic lung disease
- Presence of pneumonia
- Cardiomyopathy or congestive heart failure
- History of tachyarrhythmias, with the exception of sinus tachycardia
- HR ≥ 150 bpm
- Potassium level ≤ 3.0
- > 15 pack-year smoking history
- Fever ≥ 101.5ºF
- BP ≥ 170/100 mmHg

8

Emergency Department Procedures and Study Entry

- Study length: app. 6.5 hr (Hour -1.5 to Hour 5)
- Subjects received standardized care consistent with the National Asthma Education and Prevention Program (NAEPP) guidelines
- If FEV$_1$ ≤ 55 % of predicted after receiving SOC, subject was randomized to receive either MN-221 or placebo
- After randomization, the subject received the study drug (MN-221) or placebo
- Until the subject's FEV$_1$ reached ≥ 70% of predicted, the subject continued to receive SOC

9

Methodology

Efficacy

- The primary efficacy parameter was the change from Baseline in FEV$_1$, expressed as percent of predicted (%Pred), at Hour 5
- Secondary efficacy:
 - Albuterol use while in study
 - Hospitalization rate
 - FEV$_1$ (L) Change from Baseline (Hour 5)
 - Hospitalization Rate

Safety & PK

- Safety assessments included
 - AEs
 - Clinical laboratory testing
 - Pulse oximetry
 - Physical examinations
 - 12-lead ECG and Holter monitoring
 - Vital signs
 - CMs
- MN-221 and albuterol PK and PD analyses

10

Study Design & Schedule

- Randomized, placebo-controlled, single-blind, dose escalation
- 29 patients with acute exacerbations of asthma (FEV$_1$ ≤ 55% of predicted) at 8 ED sites
- Doses:
 - 16 µg/min for 15 min (total dose of 240 µg)
 - 30 µg/min for 15 min (total dose of 450 µg)
 - 16 µg/min for 15 min + 8 µg/min for 105 min (total dose of 1,080 µg)
 - 2 patients received total dose of 1,995 µg
- Patients received Standard of Care (SOC) treatment in addition to adjunctive treatment with MN-221 or placebo
- Outcome measures – descriptive statistics only – FEV$_1$, PK, safety

11

Mean FEV$_1$ (% Predicted) Change from Baseline (Hour 5)

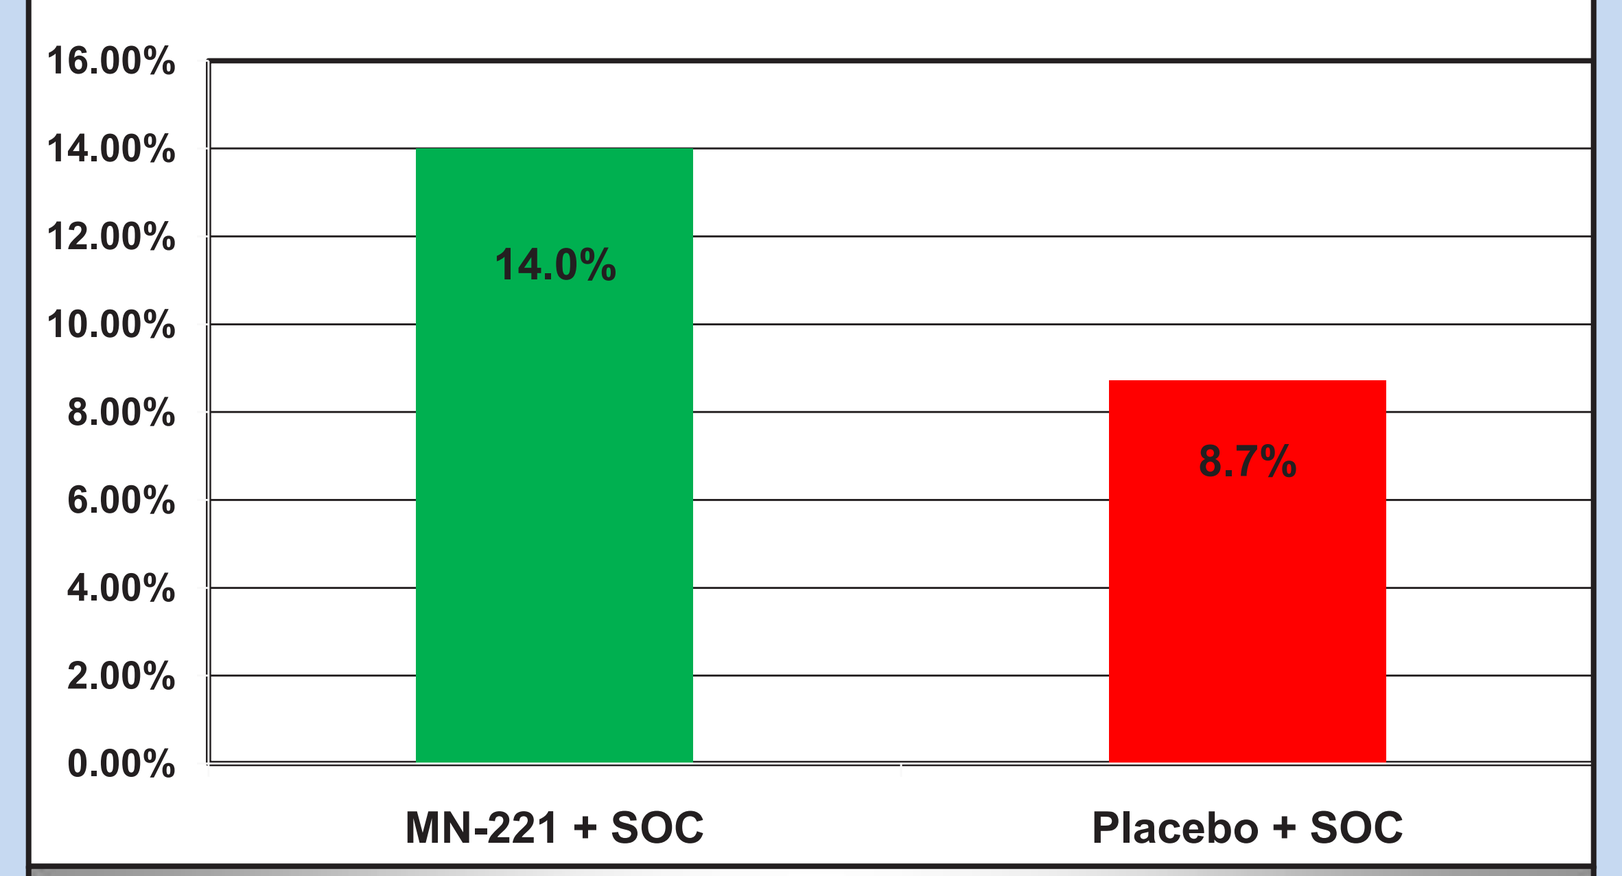
16.00%
14.00%
12.00%
10.00%
8.00%
6.00%
4.00%
2.00%
0.00%
14.0%
8.7%
MN-221 + SOC
Placebo + SOC

Mean FEV$_1$ (% Predicted) Change from Baseline was 5.3% higher in the MN-221 dose group vs. the Placebo group

12

Mean FEV$_1$(L) Change from Baseline (Hour 5)

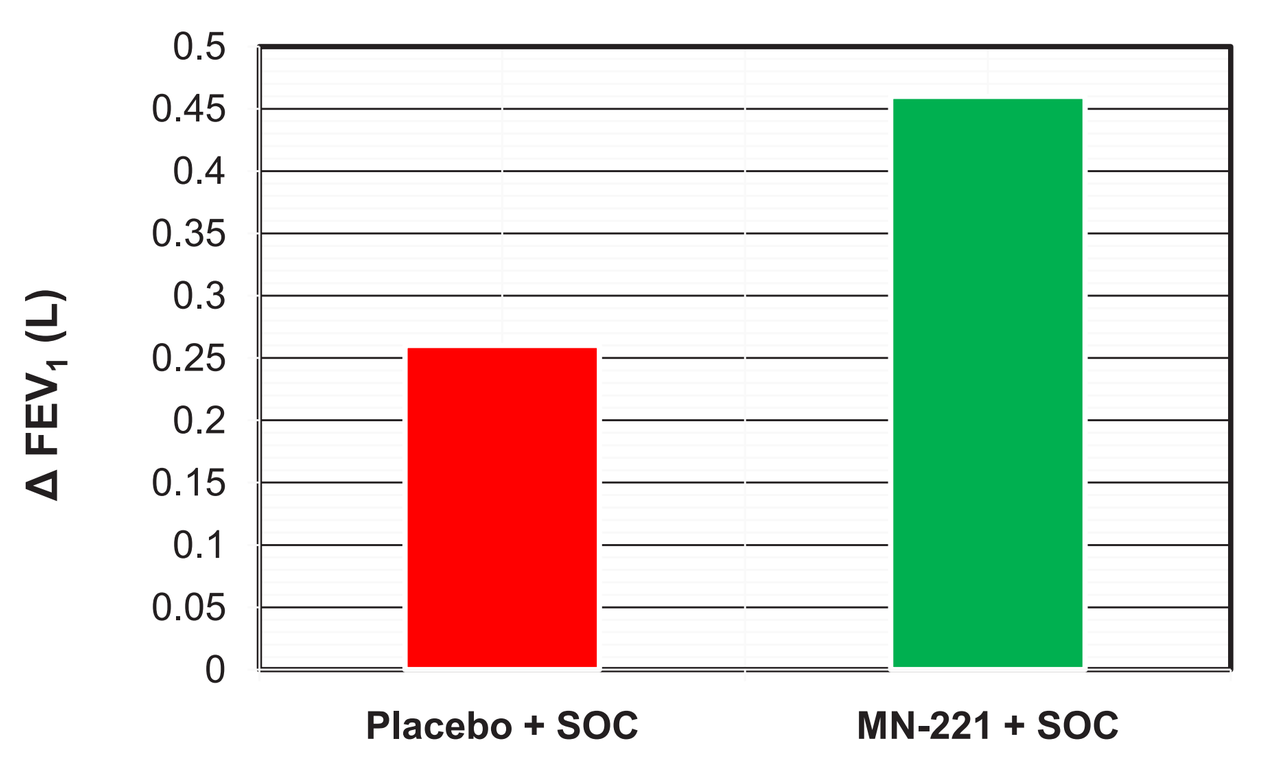
0.5
0.45
0.4
0.35
0.3
0.25
0.2
0.15
0.1
0.05
0
Δ FEV$_1$ (L)
Placebo + SOC
MN-221 + SOC

Mean FEV$_1$(L) Change from Baseline was 200 mL greater in the MN-221 dose group versus the placebo group

13

Mean FEV$_1$ (% Predicted) at Hour 5 in Hospitalized vs. NOT Hospitalized Patients

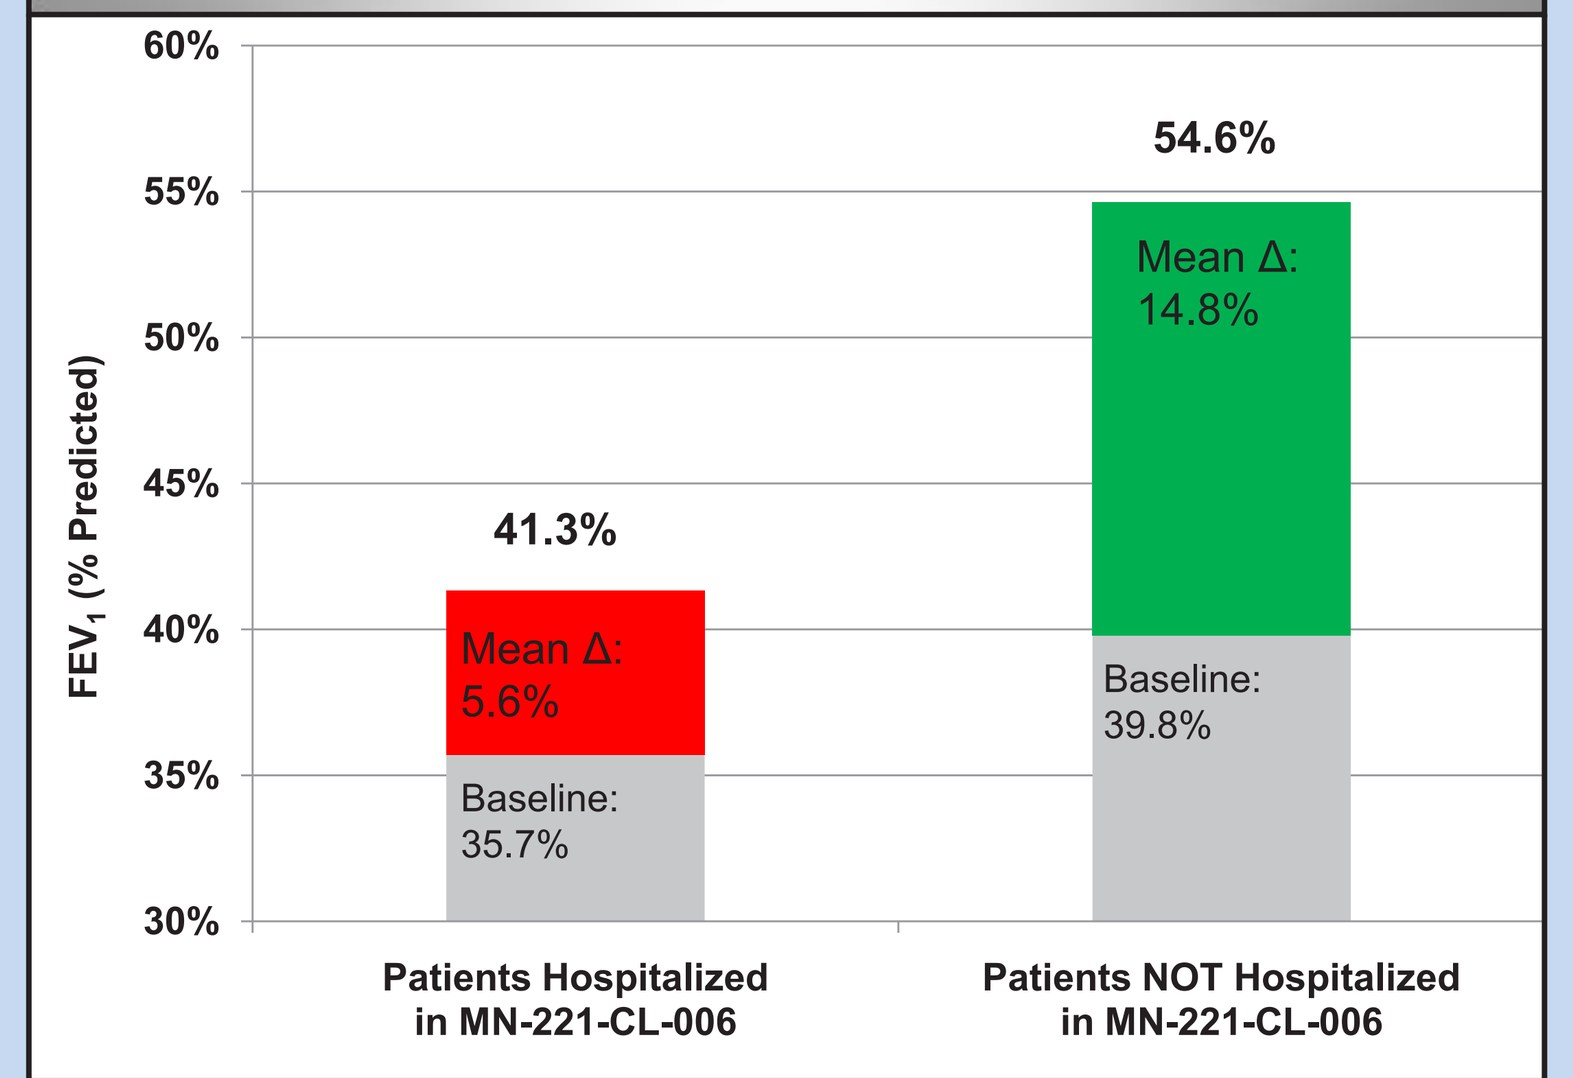
60%
55%
50%
45%
40%
35%
30%
FEV$_1$ (% Predicted)
41.3%
Mean Δ: 5.6%
Baseline: 35.7%
54.6%
Mean Δ: 14.8%
Baseline: 39.8%
Patients Hospitalized in MN-221-CL-006
Patients NOT Hospitalized in MN-221-CL-006

14

Treatment Period Heart Rate Change

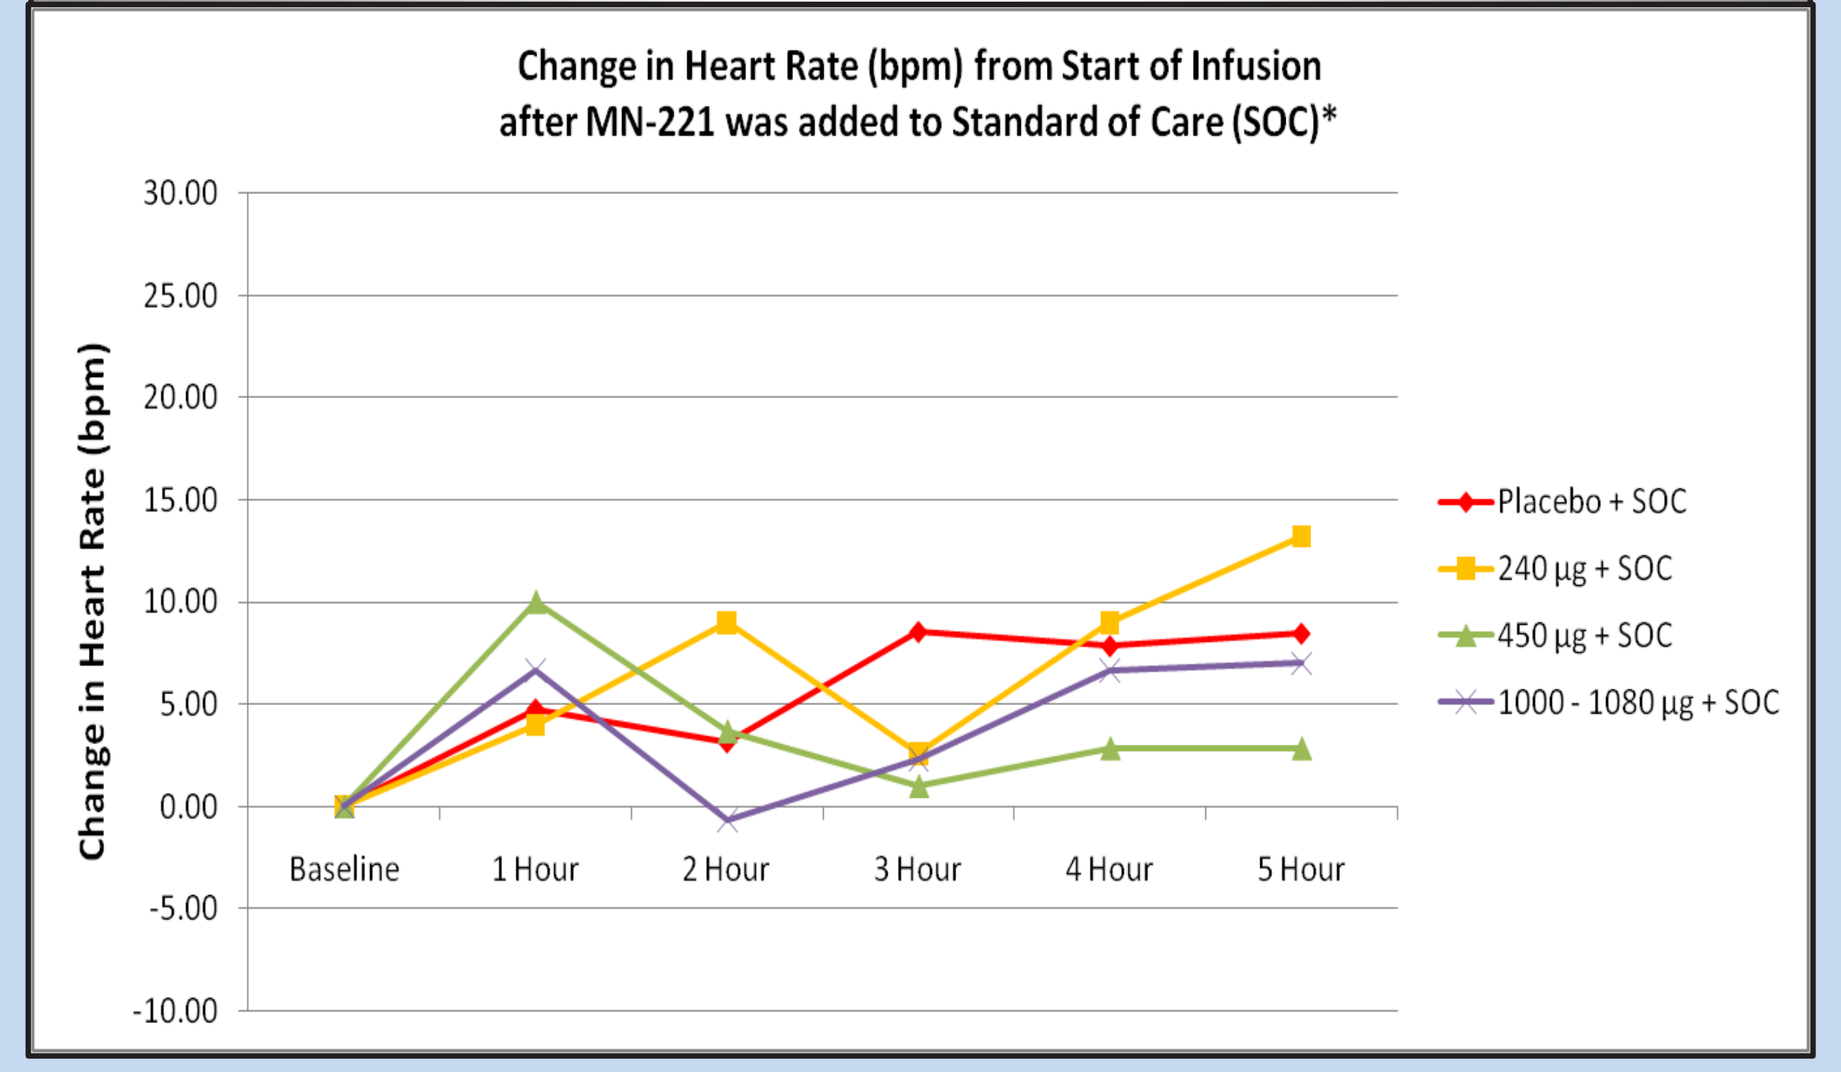
Change in Heart Rate (bpm) from Start of Infusion after MN-221 was added to Standard of Care (SOC)*
30.00
25.00
20.00
15.00
10.00
5.00
0.00
-5.00
-10.00
Change in Heart Rate (bpm)
Baseline
1 Hour
2 Hour
3 Hour
4 Hour
5 Hour
Placebo + SOC
240 µg + SOC
450 µg + SOC
1000 - 1080 µg + SOC

* All patients received Standard of Care (SOC) including inhaled albuterol, inhaled ipratropium, and steroids.

15

Summary of Adverse Events

- No deaths.
- 2 (2/16, 13%) subjects in the all MN-221 group and 5 (5/13, 39%) in the placebo group experienced a total of 8 treatment-emergent SAEs. All events resolved. No discontinuations due to an AE.
- Both SAE subjects in the MN-221 group experienced a single SAE of asthma. 5 Placebo group SAEs were events of asthma and 1 subject also experienced moderate pneumonia considered not related to study drug.
- Among 16 subjects in the all MN-221 group, 6 (38%) experienced at least one TEAE compared with 6 of 13 (46%) subjects in the placebo group. Majority were mild or moderate in intensity. One subject who received 1995 µg of MN-221 experienced a severe TEAE (asthma) that resolved.
- No observed clinically significant abnormal ECG findings.

16

Average (± SD) Concentrations of MN-221 in Plasma (Linear Scale) after Intravenous Infusion

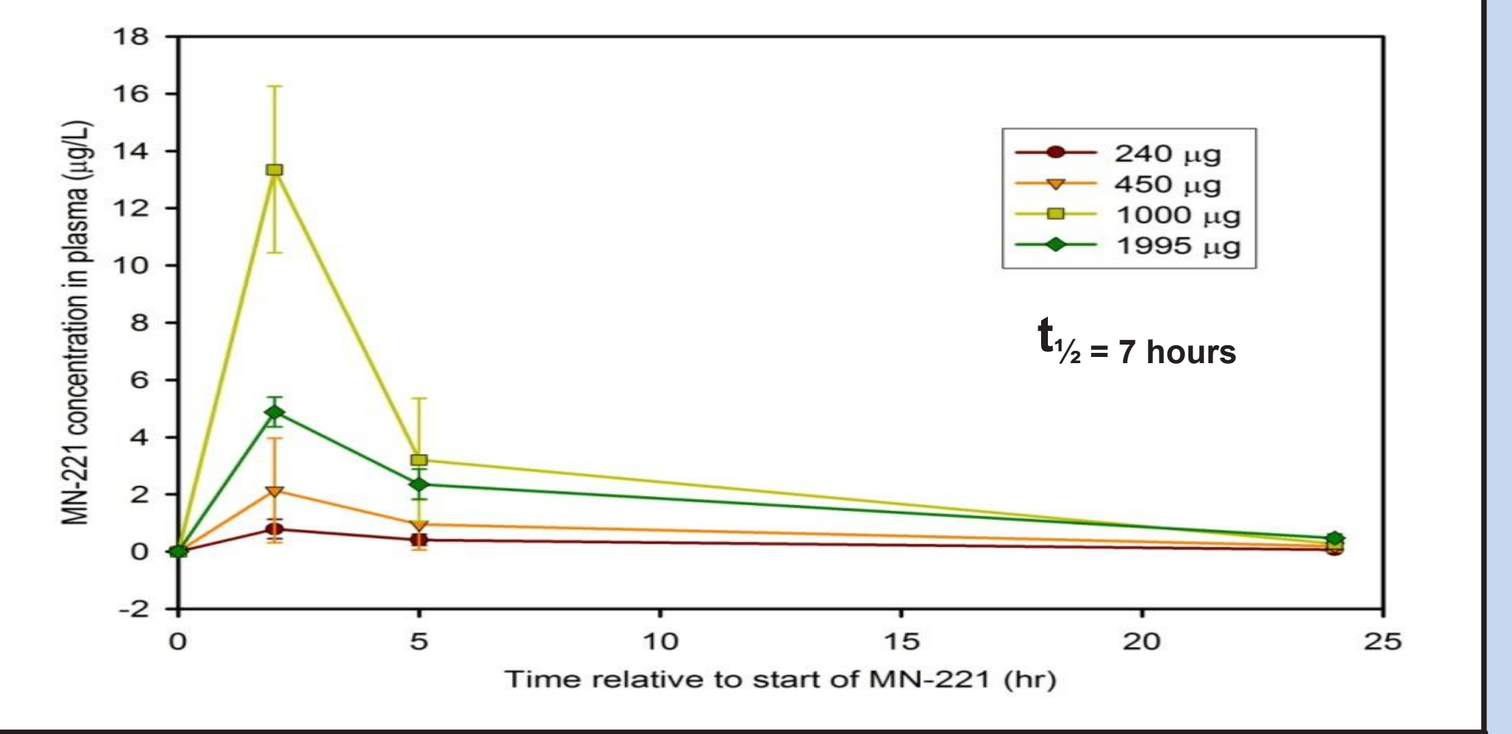
18
16
14
12
10
8
6
4
2
0
-2
MN-221 concentration in plasma (µg/L)
0
5
10
15
20
25
Time relative to start of MN-221 (hr)
240 µg
450 µg
1000 µg
1995 µg
t$_{½}$ = 7 hours

Average concentrations declined in parallel for the 240, 450, and 1995 µg groups.
1000 µg group: 7-hr delay in 24-hour sample collection from 1 subject.
Average concentrations dose-proportional between the 240 and 450 µg doses at all sampling times. Two subjects received the higher dose.

17

Conclusions and Clinical Implications

- MN-221 was well tolerated and demonstrated efficacy as measured by improvements in FEV$_1$
- There were no safety concerns with adding MN-221 to the standard of care.
- There was a reduction in the hospitalization rate among patients treated with MN-221.
- The data support further investigation of the use of MN-221 as an adjunctive treatment for subjects with an acute exacerbation of asthma